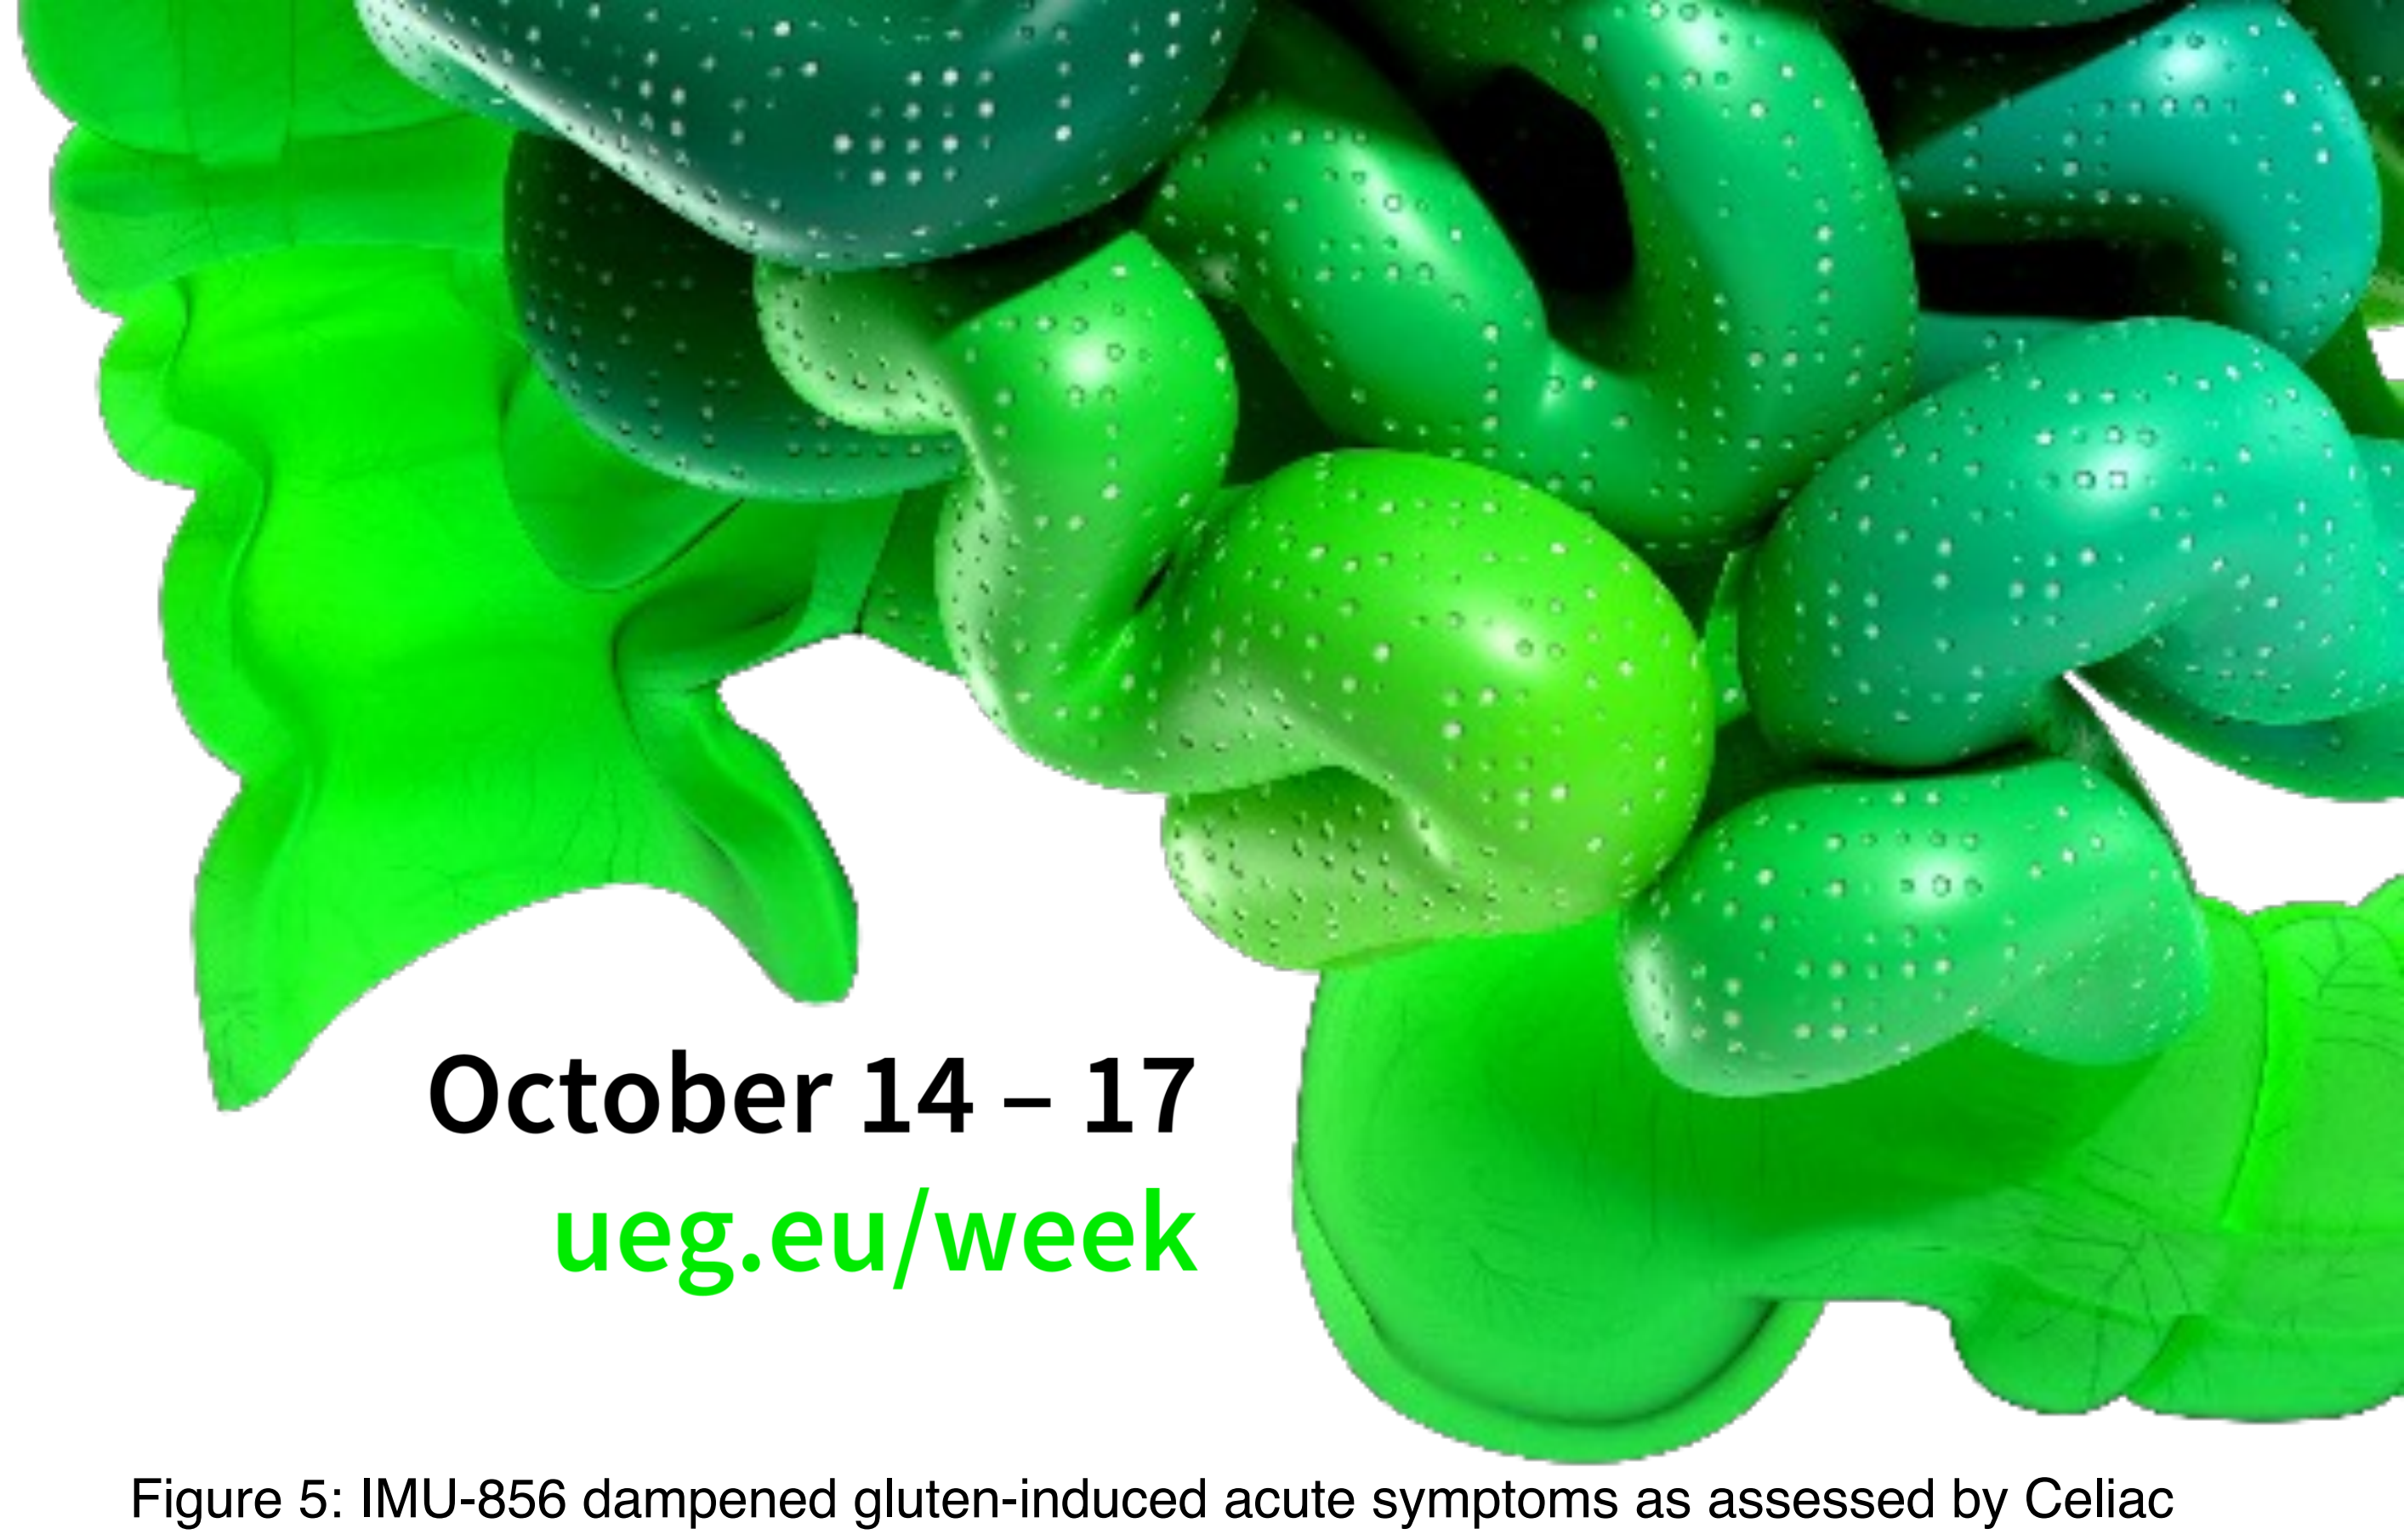
October 14 – 17
ueg.eu/week

FIRST IN HUMAN TRIAL OF IMU-856, AN ORALLY AVAILABLE EPIGENETIC MODULATOR OF BARRIER REGENERATION FOR THE TREATMENT OF CELIAC DISEASE

A. James M. Daveson[1], Franziska Buriánek[2], Jelena Mihajlović[3], Evelyn Peelen[2], Juliano Fonseca[2], Amelie Schreieck[2], Martina Wirth[2], Inge Kehler[2], Daniel Vitt[2], Hella Kohlhof[2], Andreas Muehler[2]

[1] Wesley Medical Research and Coral Sea Clinical Research Institute, QLD, Australia
[2] Immunic AG, Gräfelfing, Germany
[3] Formerly Immunic AG, Gräfelfing, Germany

Mechanism of action of IMU-856

- IMU-856 is a highly selective and potent modulator of the epigenetic regulator SIRT6 (Sirtuin 6)
- IMU-856 improves quality and function of bowel epithelium through activation of physiological (gut) regeneration
- IMU-856 enhances intestinal barrier function by modulating tight junction proteins
- IMU-856 has no known immuno-suppressive effects

Overview of IMU-856 phase 1b clinical trial in celiac disease

Figure 1: Flow chart of IMU-856 double-blind, randomized, placebo controlled Phase 1b trial.

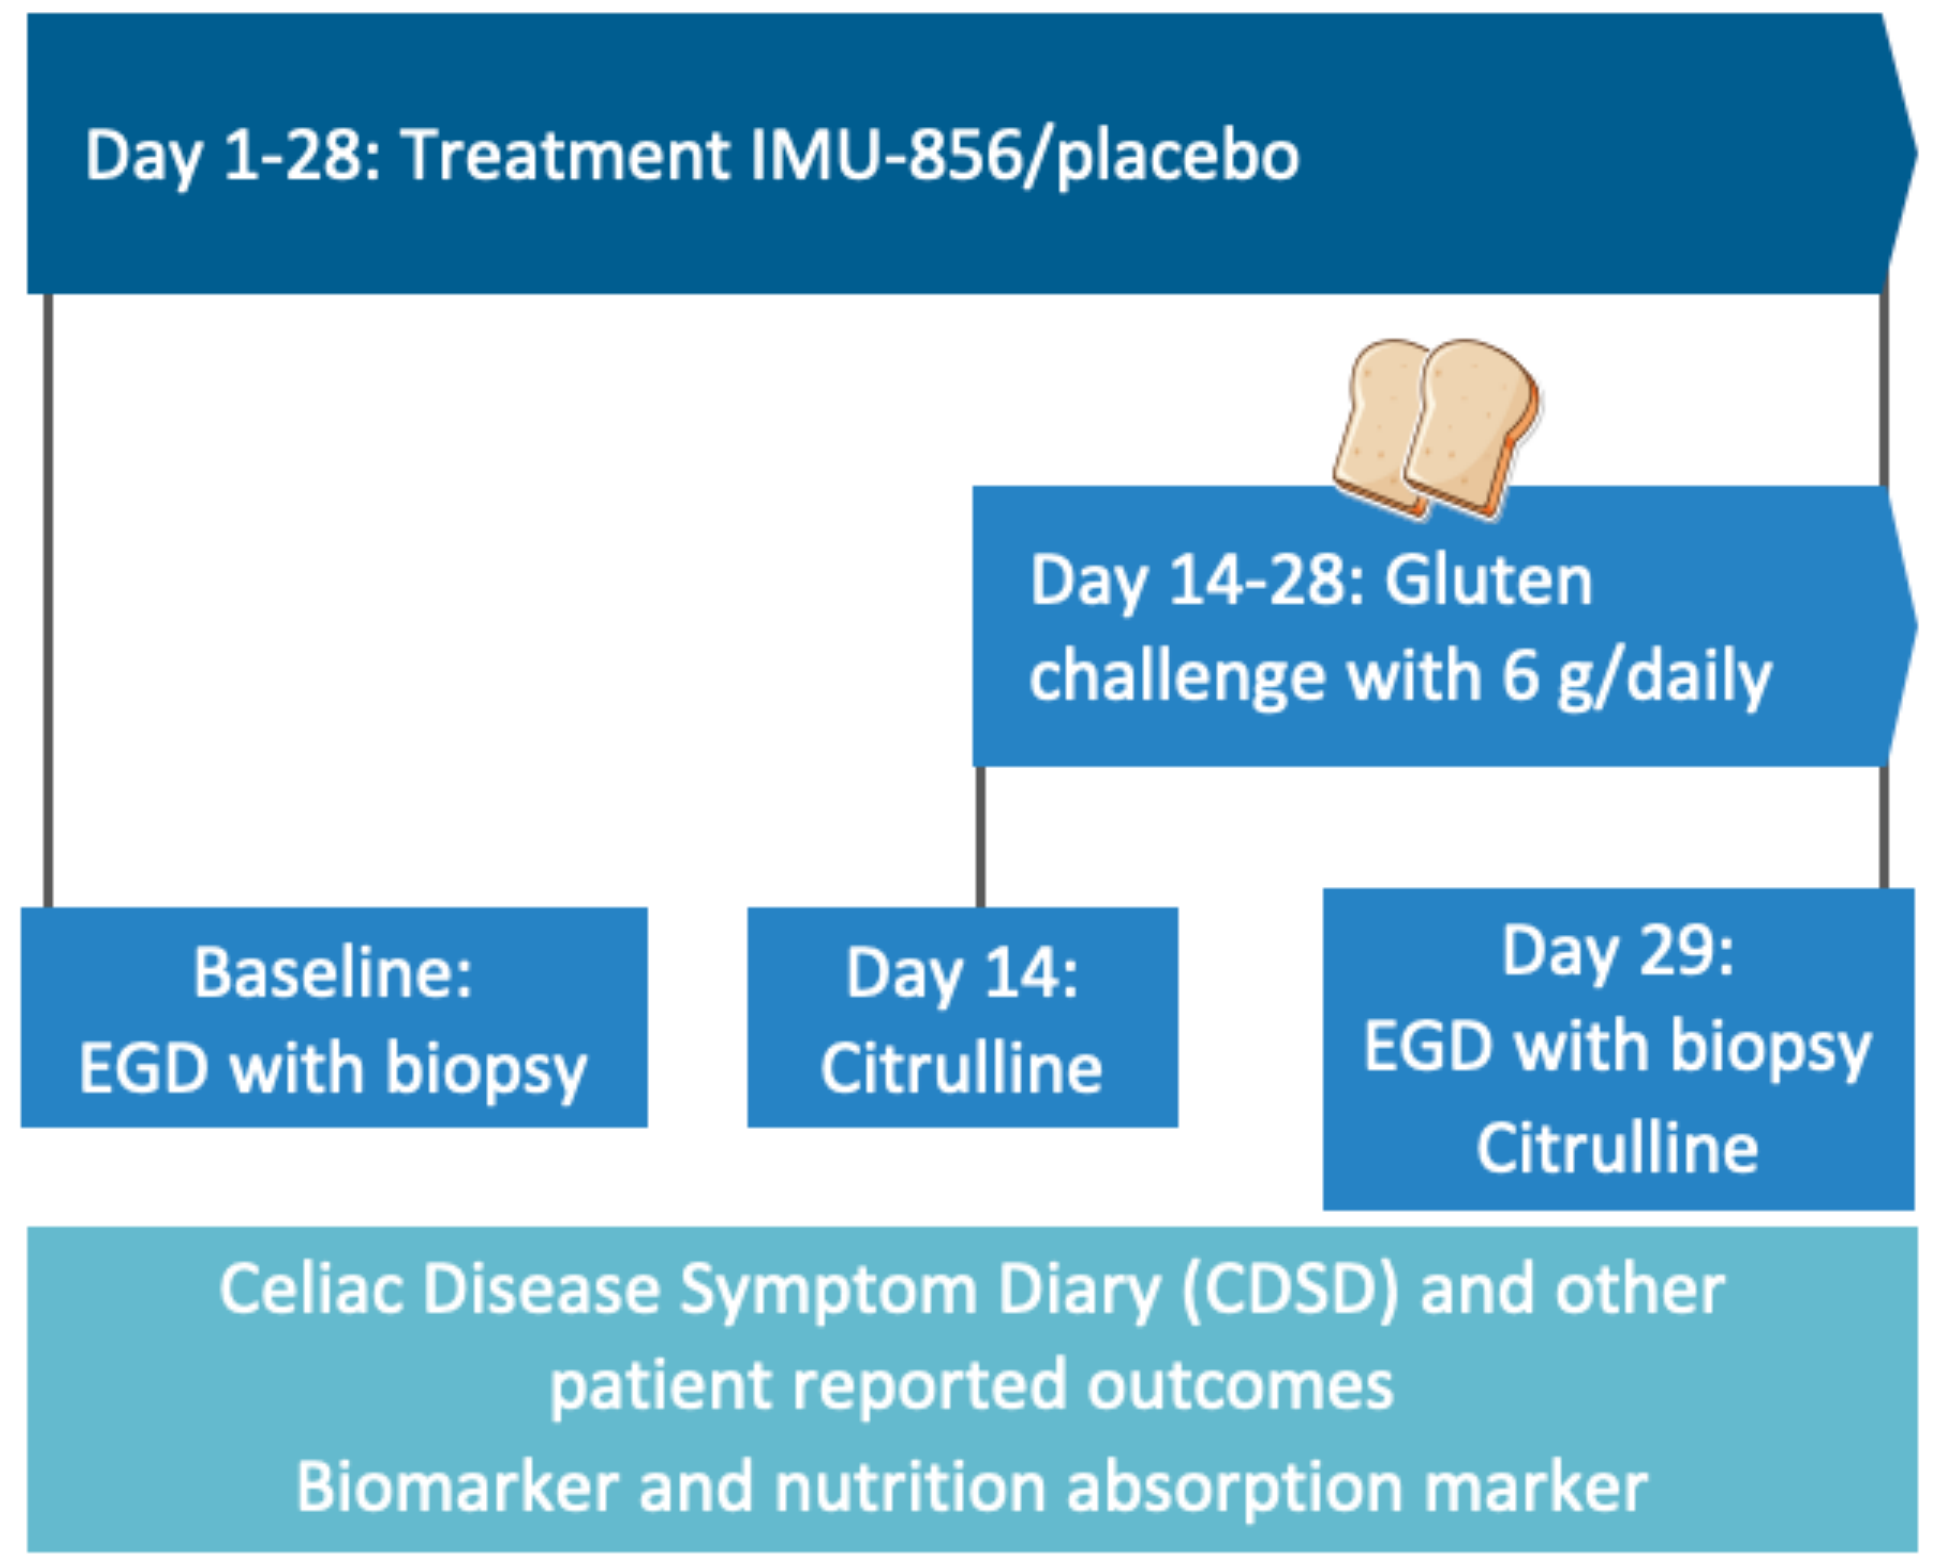
Day 1-28: Treatment IMU-856/placebo
Day 14-28: Gluten challenge with 6 g/daily
Baseline: EGD with biopsy
Day 14: Citrulline
Day 29: EGD with biopsy Citrulline
Celiac Disease Symptom Diary (CDSD) and other patient reported outcomes
Biomarker and nutrition absorption marker

- IMU-856 Phase 1b trial designed to include a well-controlled celiac disease patient population
- 28 days of dosing (80mg or 160mg IMU-856 QD)
- 15 days of gluten challenge (6g gluten/daily)
- N enrolled/completed = 43/35 patients (IMU-856: N=29/24)
- Performed at sites in Australia and New Zealand
- Designed to assess safety, tolerability, pharmacodynamics and pharmacokinetics of IMU-856
- Measured histologic changes, blood bio-markers, nutrient uptake and disease-related symptoms

Proof-of-concept study: positive results from IMU-856 phase 1b clinical trial

Figure 2: IMU-856 protected against gluten-induced intestinal epithelial damage by substantially and significantly reducing the decrease in villous height as compared to placebo.

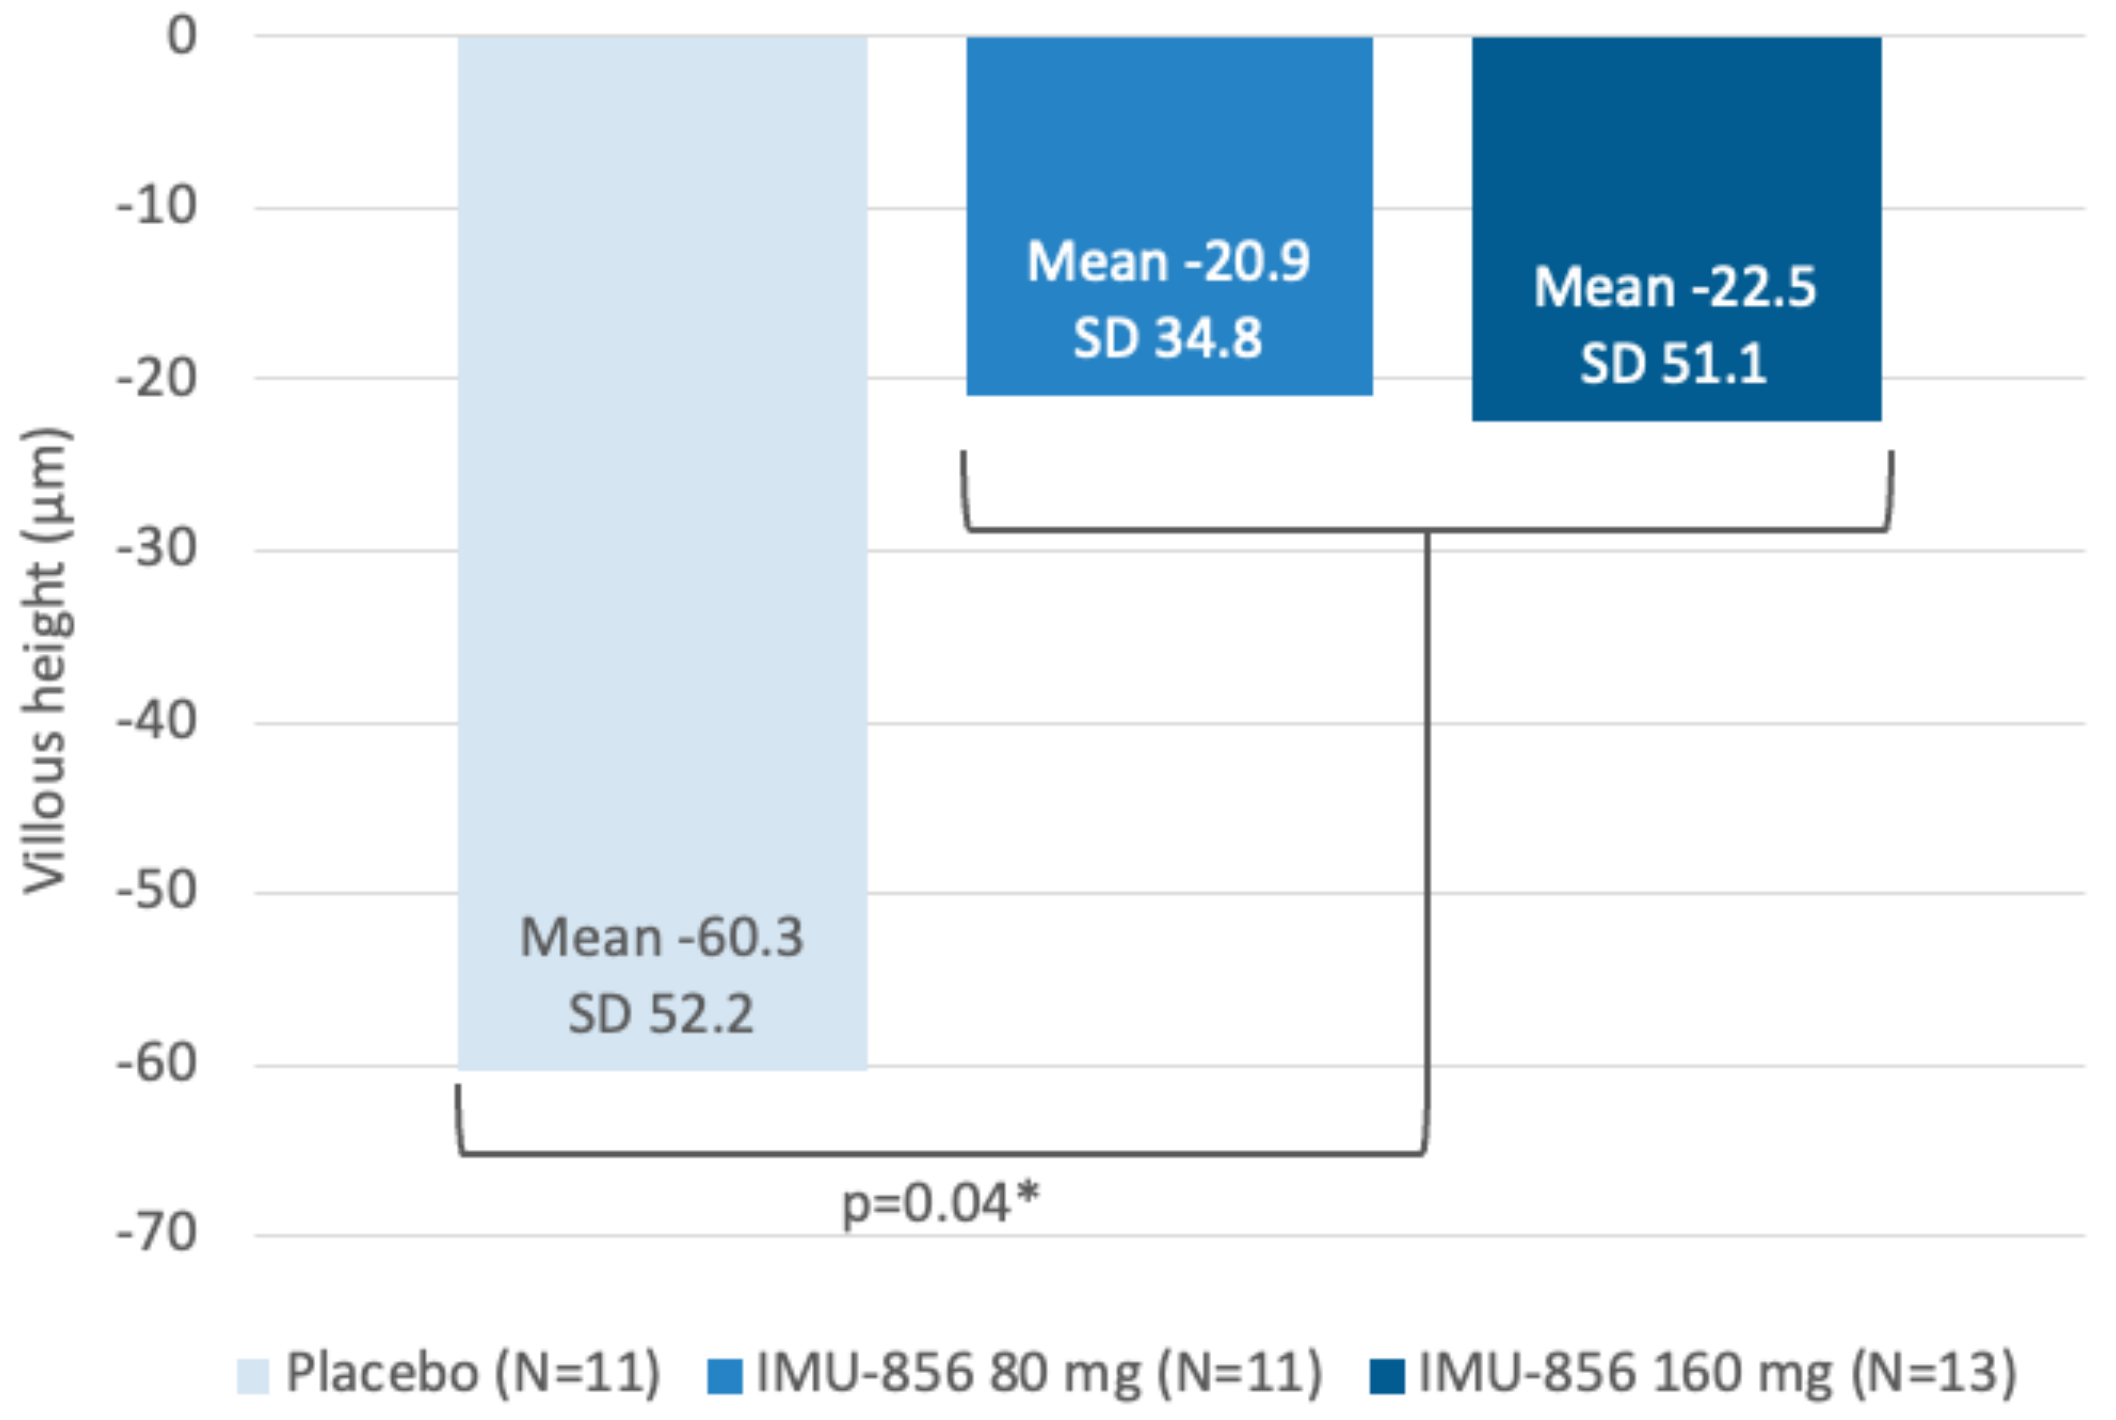
Villous height (µm)
0
-10
-20
-30
-40
-50
-60
-70
Mean -60.3
SD 52.2
Mean -20.9
SD 34.8
Mean -22.5
SD 51.1
p=0.04*
Placebo (N=11)
IMU-856 80 mg (N=11)
IMU-856 160 mg (N=13)

- *Wilcoxon Two-Sample Test comparison between pooled IMU-856 groups and placebo, performed as post-hoc exploratory statistical analysis.*

Figure 3: IMU-856 improved plasma citrulline levels (biomarker for enterocyte health) already within the first 2 weeks prior to gluten challenge. This improvement was further maintained throughout the trial including a 15-day gluten challenge.

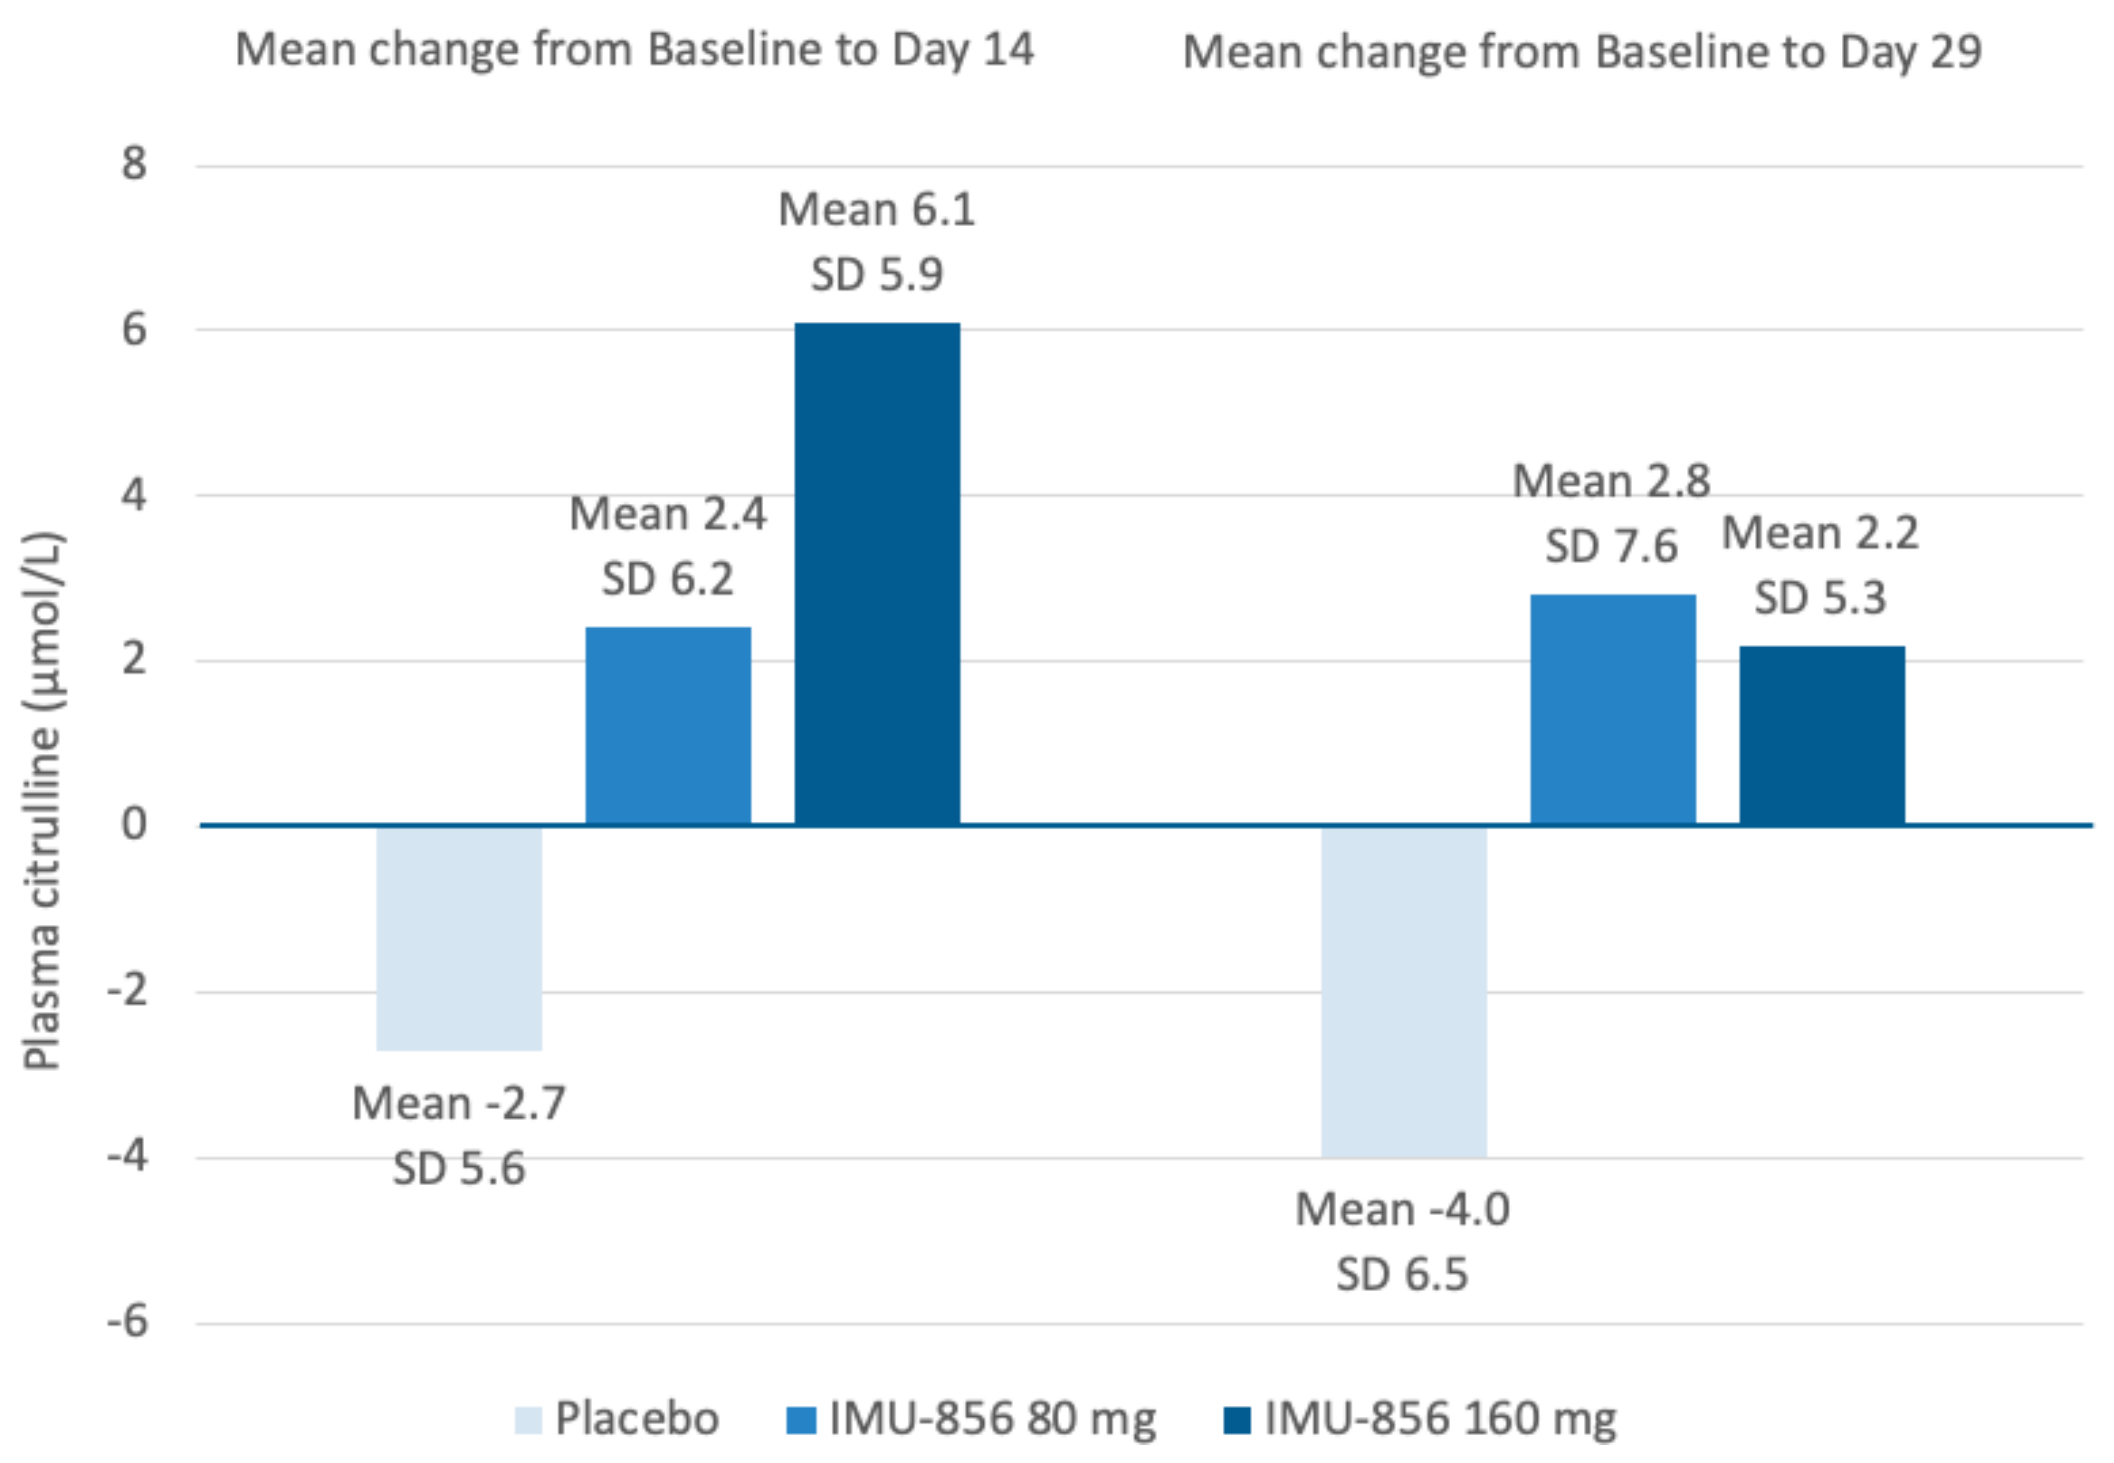
Mean change from Baseline to Day 14
Mean change from Baseline to Day 29
Plasma citrulline (µmol/L)
8
6
4
2
0
-2
-4
-6
Mean -2.7
SD 5.6
Mean 2.4
SD 6.2
Mean 6.1
SD 5.9
Mean -4.0
SD 6.5
Mean 2.8
SD 7.6
Mean 2.2
SD 5.3
Placebo
IMU-856 80 mg
IMU-856 160 mg

Number of Patients: Placebo: N=13 for Mean Change Baseline to Day 14, N=11 for Mean Change Baseline to Day 29; IMU-856 80 mg: N=14 for Mean Change Baseline to Day 14, N=11 for Mean Change Baseline to Day 29; IMU-856 160 mg: N=13 for Mean Change Baseline to Day 14, N=13 for Mean Change Baseline to Day 29.

Figure 4: IMU-856 showed enhanced nutrient absorption as exemplified by Vitamin B12 levels.

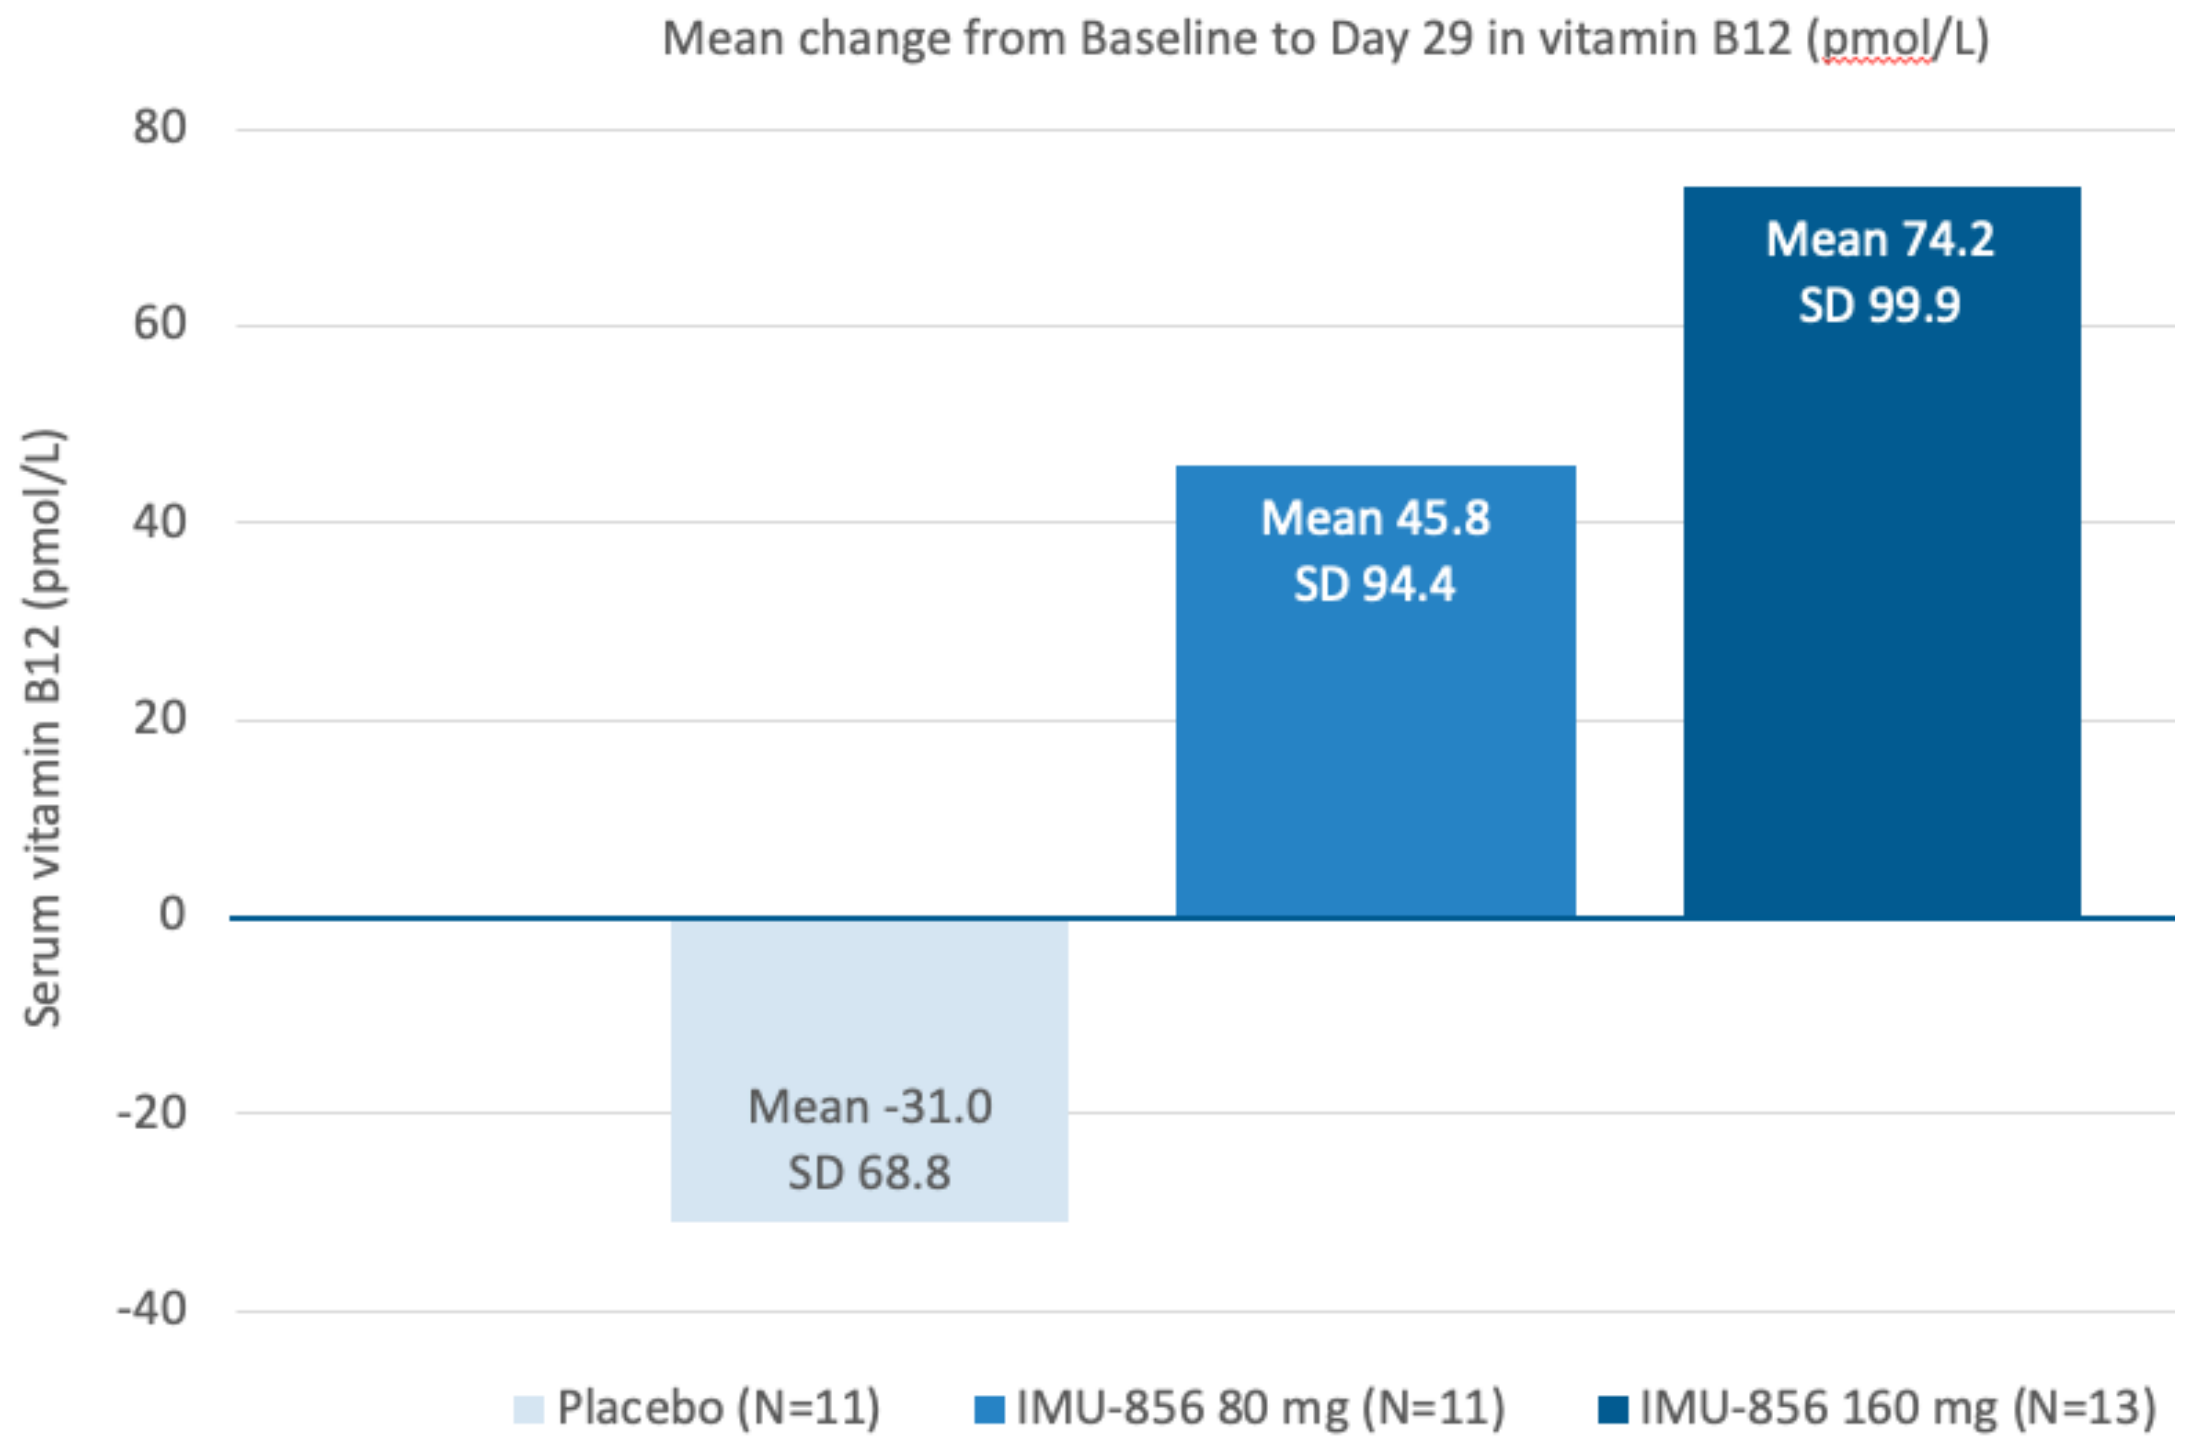
Mean change from Baseline to Day 29 in vitamin B12 (pmol/L)
Serum vitamin B12 (pmol/L)
80
60
40
20
0
-20
-40
Mean -31.0
SD 68.8
Mean 45.8
SD 94.4
Mean 74.2
SD 99.9
Placebo (N=11)
IMU-856 80 mg (N=11)
IMU-856 160 mg (N=13)

Figure 5: IMU-856 dampened gluten-induced acute symptoms as assessed by Celiac Disease Symptom Diary (CDSD).

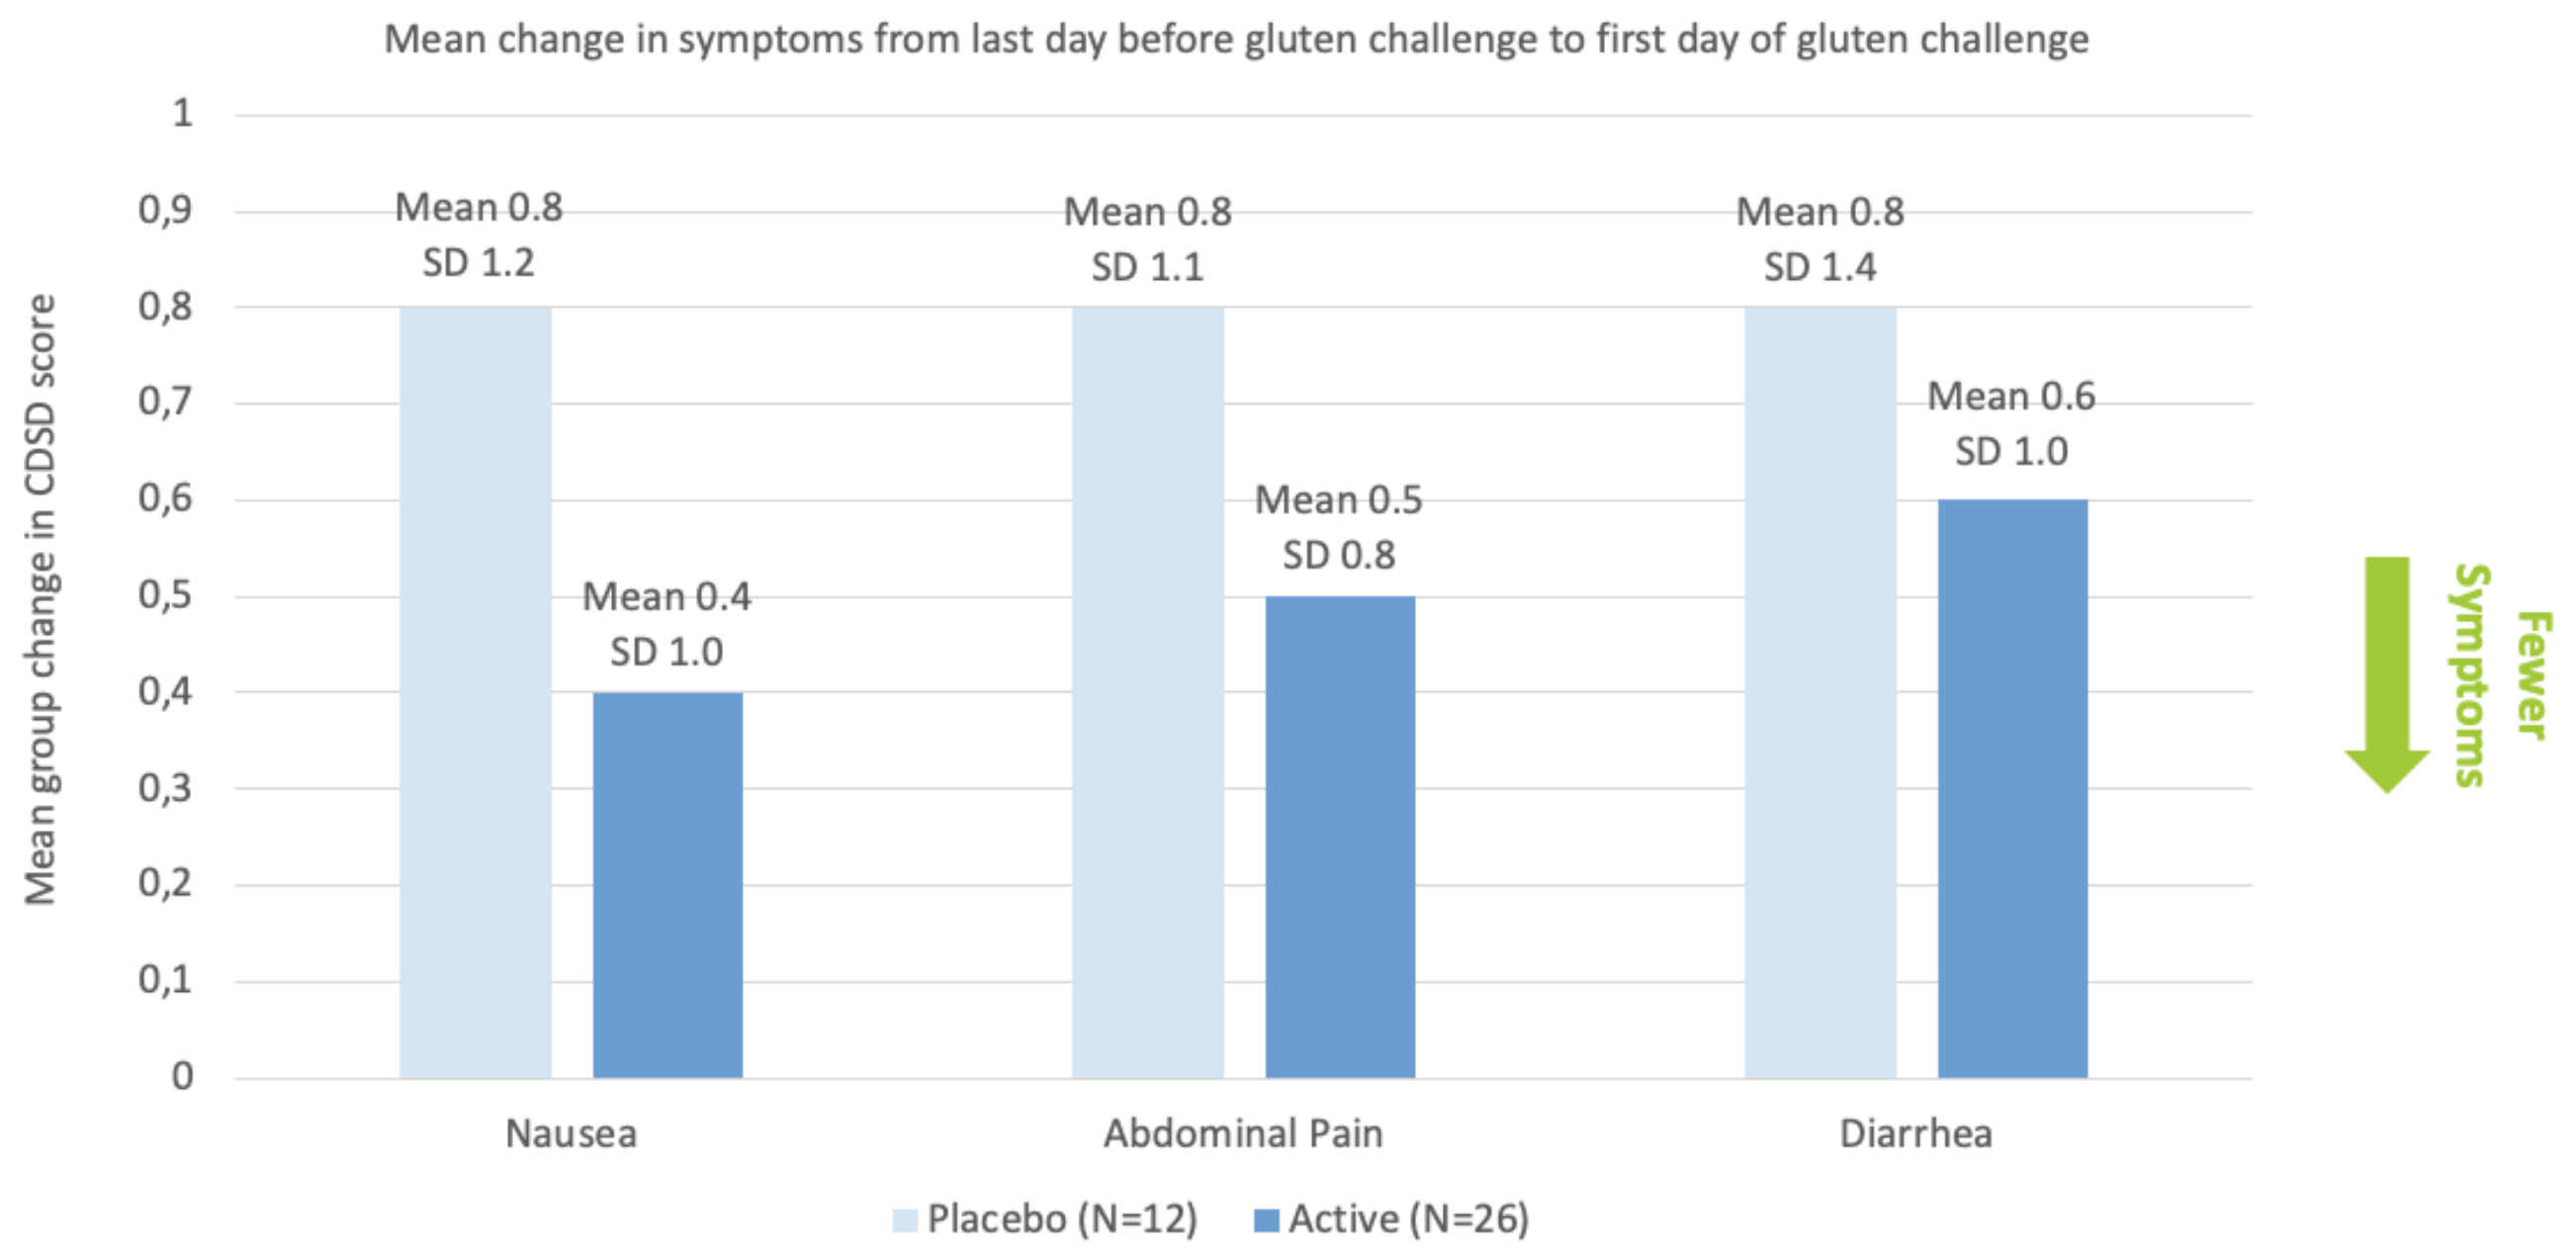
Mean change in symptoms from last day before gluten challenge to first day of gluten challenge
Mean group change in CDSD score
1
0,9
0,8
0,7
0,6
0,5
0,4
0,3
0,2
0,1
0
Mean 0.8
SD 1.2
Mean 0.4
SD 1.0
Mean 0.8
SD 1.1
Mean 0.5
SD 0.8
Mean 0.8
SD 1.4
Mean 0.6
SD 1.0
Nausea
Abdominal Pain
Diarrhea
Placebo (N=12)
Active (N=26)
Fewer Symptoms

Safety Summary of IMU-856

IMU-856 shown to be safe and well-tolerated

- No dose-dependency in adverse events
- No IMP-related severe treatment-emergent adverse events
- No IMP-related serious adverse events
- No trend in post-dose changes in laboratory parameters
- No clinically relevant changes in vital signs, physical examination or ECG parameters

Conclusions

Figure 6: IMU-856 showed positive effects in the main four dimensions of clinical outcome.

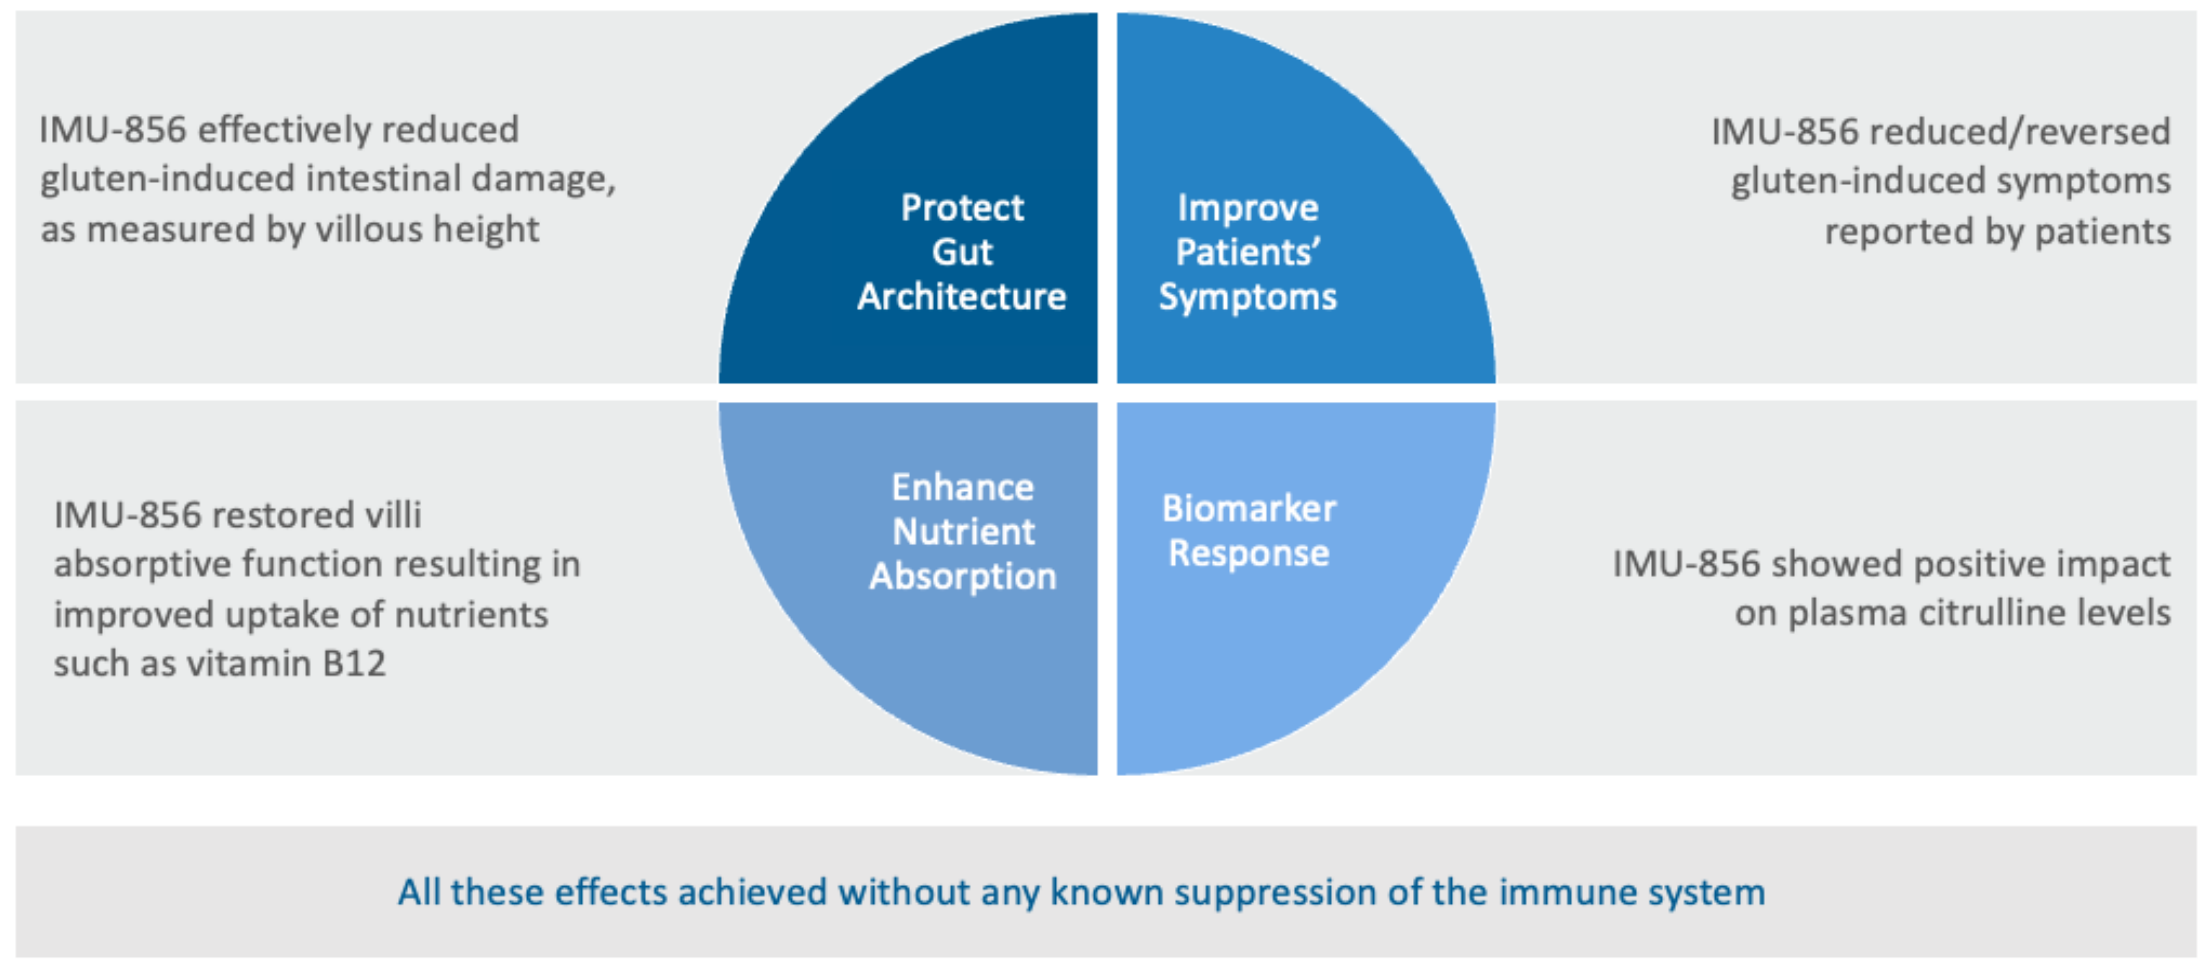
IMU-856 effectively reduced gluten-induced intestinal damage, as measured by villous height
Protect Gut Architecture
Improve Patients' Symptoms
IMU-856 reduced/reversed gluten-induced symptoms reported by patients
IMU-856 restored villi absorptive function resulting in improved uptake of nutrients such as vitamin B12
Enhance Nutrient Absorption
Biomarker Response
IMU-856 showed positive impact on plasma citrulline levels
All these effects achieved without any known suppression of the immune system

Abbr.: EGD: esophagogastroduodenoscopy; QD: quaque die = once-daily; SD: standard deviation; IMP: investigational medicinal product; ECG: electrocardiogram

Copyright © 2023 Franziska Buriánek, Senior Medical Director at Immunic AG **franziska.burianek@imux.com**

All authors apart from James Daveson are/were employed by Immunic AG