Safety, tolerability and pharmacokinetics of single oral doses of IMU-935 in healthy volunteers: First clinical experience with an orally available small molecule inhibitor of IL-17

Thomas M Polasek, Frank Fliegert, Irina Betscheider, Manfred Groeppel, Daniel Vitt, Hella Kohlhof and Andreas Muehler

Introduction

IMU-935 is an inverse agonist of the RAR-related orphan receptor RORγt, with additional but less potent inhibition of dihydroorotate dehydrogenase (DHODH). IMU-935 inhibits the secretion of several pro-inflammatory cytokines, including interleukin (IL)-17A, IL-17F and IFN-γ, differentiation of T helper 17 (Th17) cells, proliferation of activated T cells, and allows normal thymocyte maturation. With the availability and success of IL-17 targeting antibodies, IMU-935 is designed to provide an oral medication option targeting IL-17 for plaque psoriasis patients.

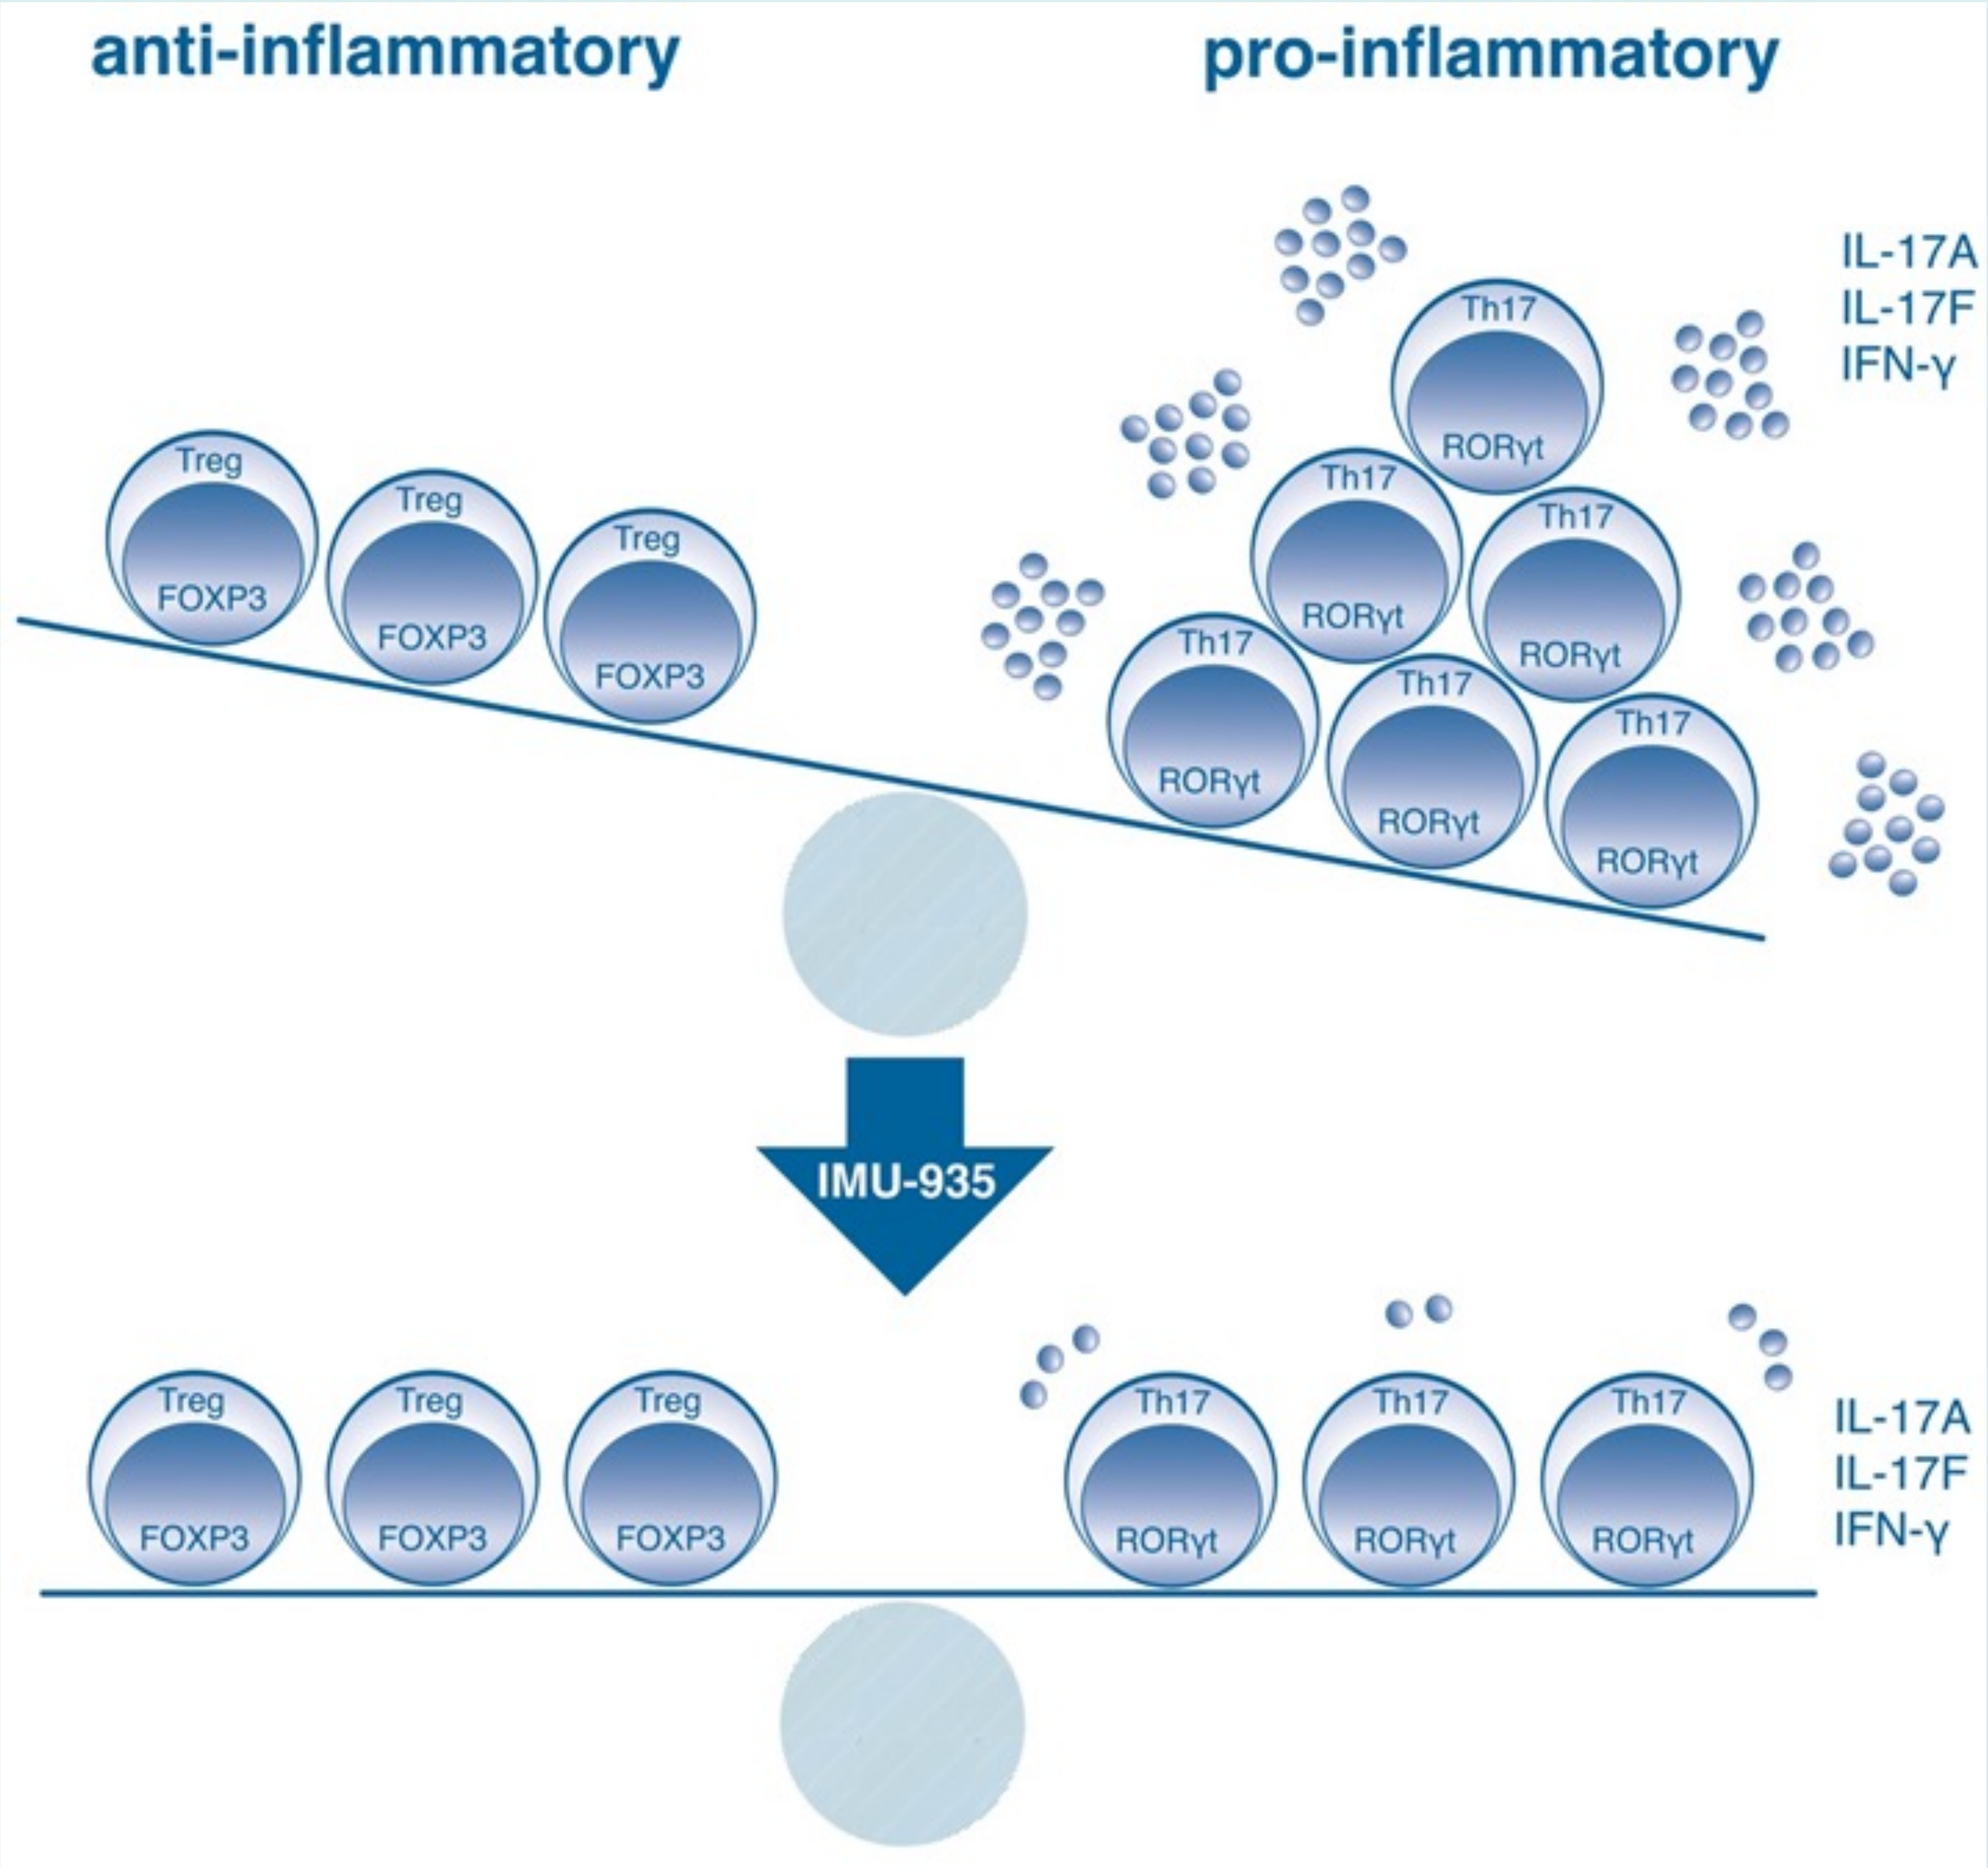
anti-inflammatory
pro-inflammatory
Treg
FOXP3
Th17
RORγt
IL-17A
IL-17F
IFN-γ
IMU-935

Figure 1 IMU-935 leads to a restoration of the balance between regulatory T cells and Th17 cells in an autoimmune setting

Methods

This is a first-in-human, double-blind, randomized, placebo-controlled clinical trial comprising three parts. In part A, healthy volunteers in cohorts of 8 subjects each were enrolled and received single ascending doses of IMU-935 or placebo (ratio 3:1).

Results

Pharmacokinetic evaluation of an IMU-935 spray-dried formulation showed plasma concentrations in most subjects at 1 hour post-dose, T_{max} between 2 to 6 hours post-dose, half-life ranging from 16.5 to 31.0 hours and dose proportional increases of C_{max} and AUC across the investigated dose range.

Table 1 IMU-935 pharmacokinetic parameters

IMU-935 dose (number of subjects)	Median (range)	Mean (%CV)		
	T_{max} (hr)	C_{max} (ng/mL)	AUC_{last} (hr*ng/mL)	$T_{1/2}$ (hr)
100 mg (N=5)	2.00 (2.00-3.00)	119 (38)	687 (25)	26.05 (27)
200 mg (N=6)	3.00 (2.00-4.00)	195 (95)	1290 (67)	24.38 (31)
300 mg (N=6)	3.50 (2.00-6.00)	182 (33)	1620 (42)	16.49 (27)
400 mg (N=6)	2.00 (2.00-6.00)	479 (27)	2530 (15)	31.00 (38)

Abbreviations: T_{max} = Time to maximum plasma concentration; C_{max} = Maximum plasma concentration; AUC_{last} = Area under the concentration time curve from 0 to the last time point; $T_{1/2}$ = Half-life

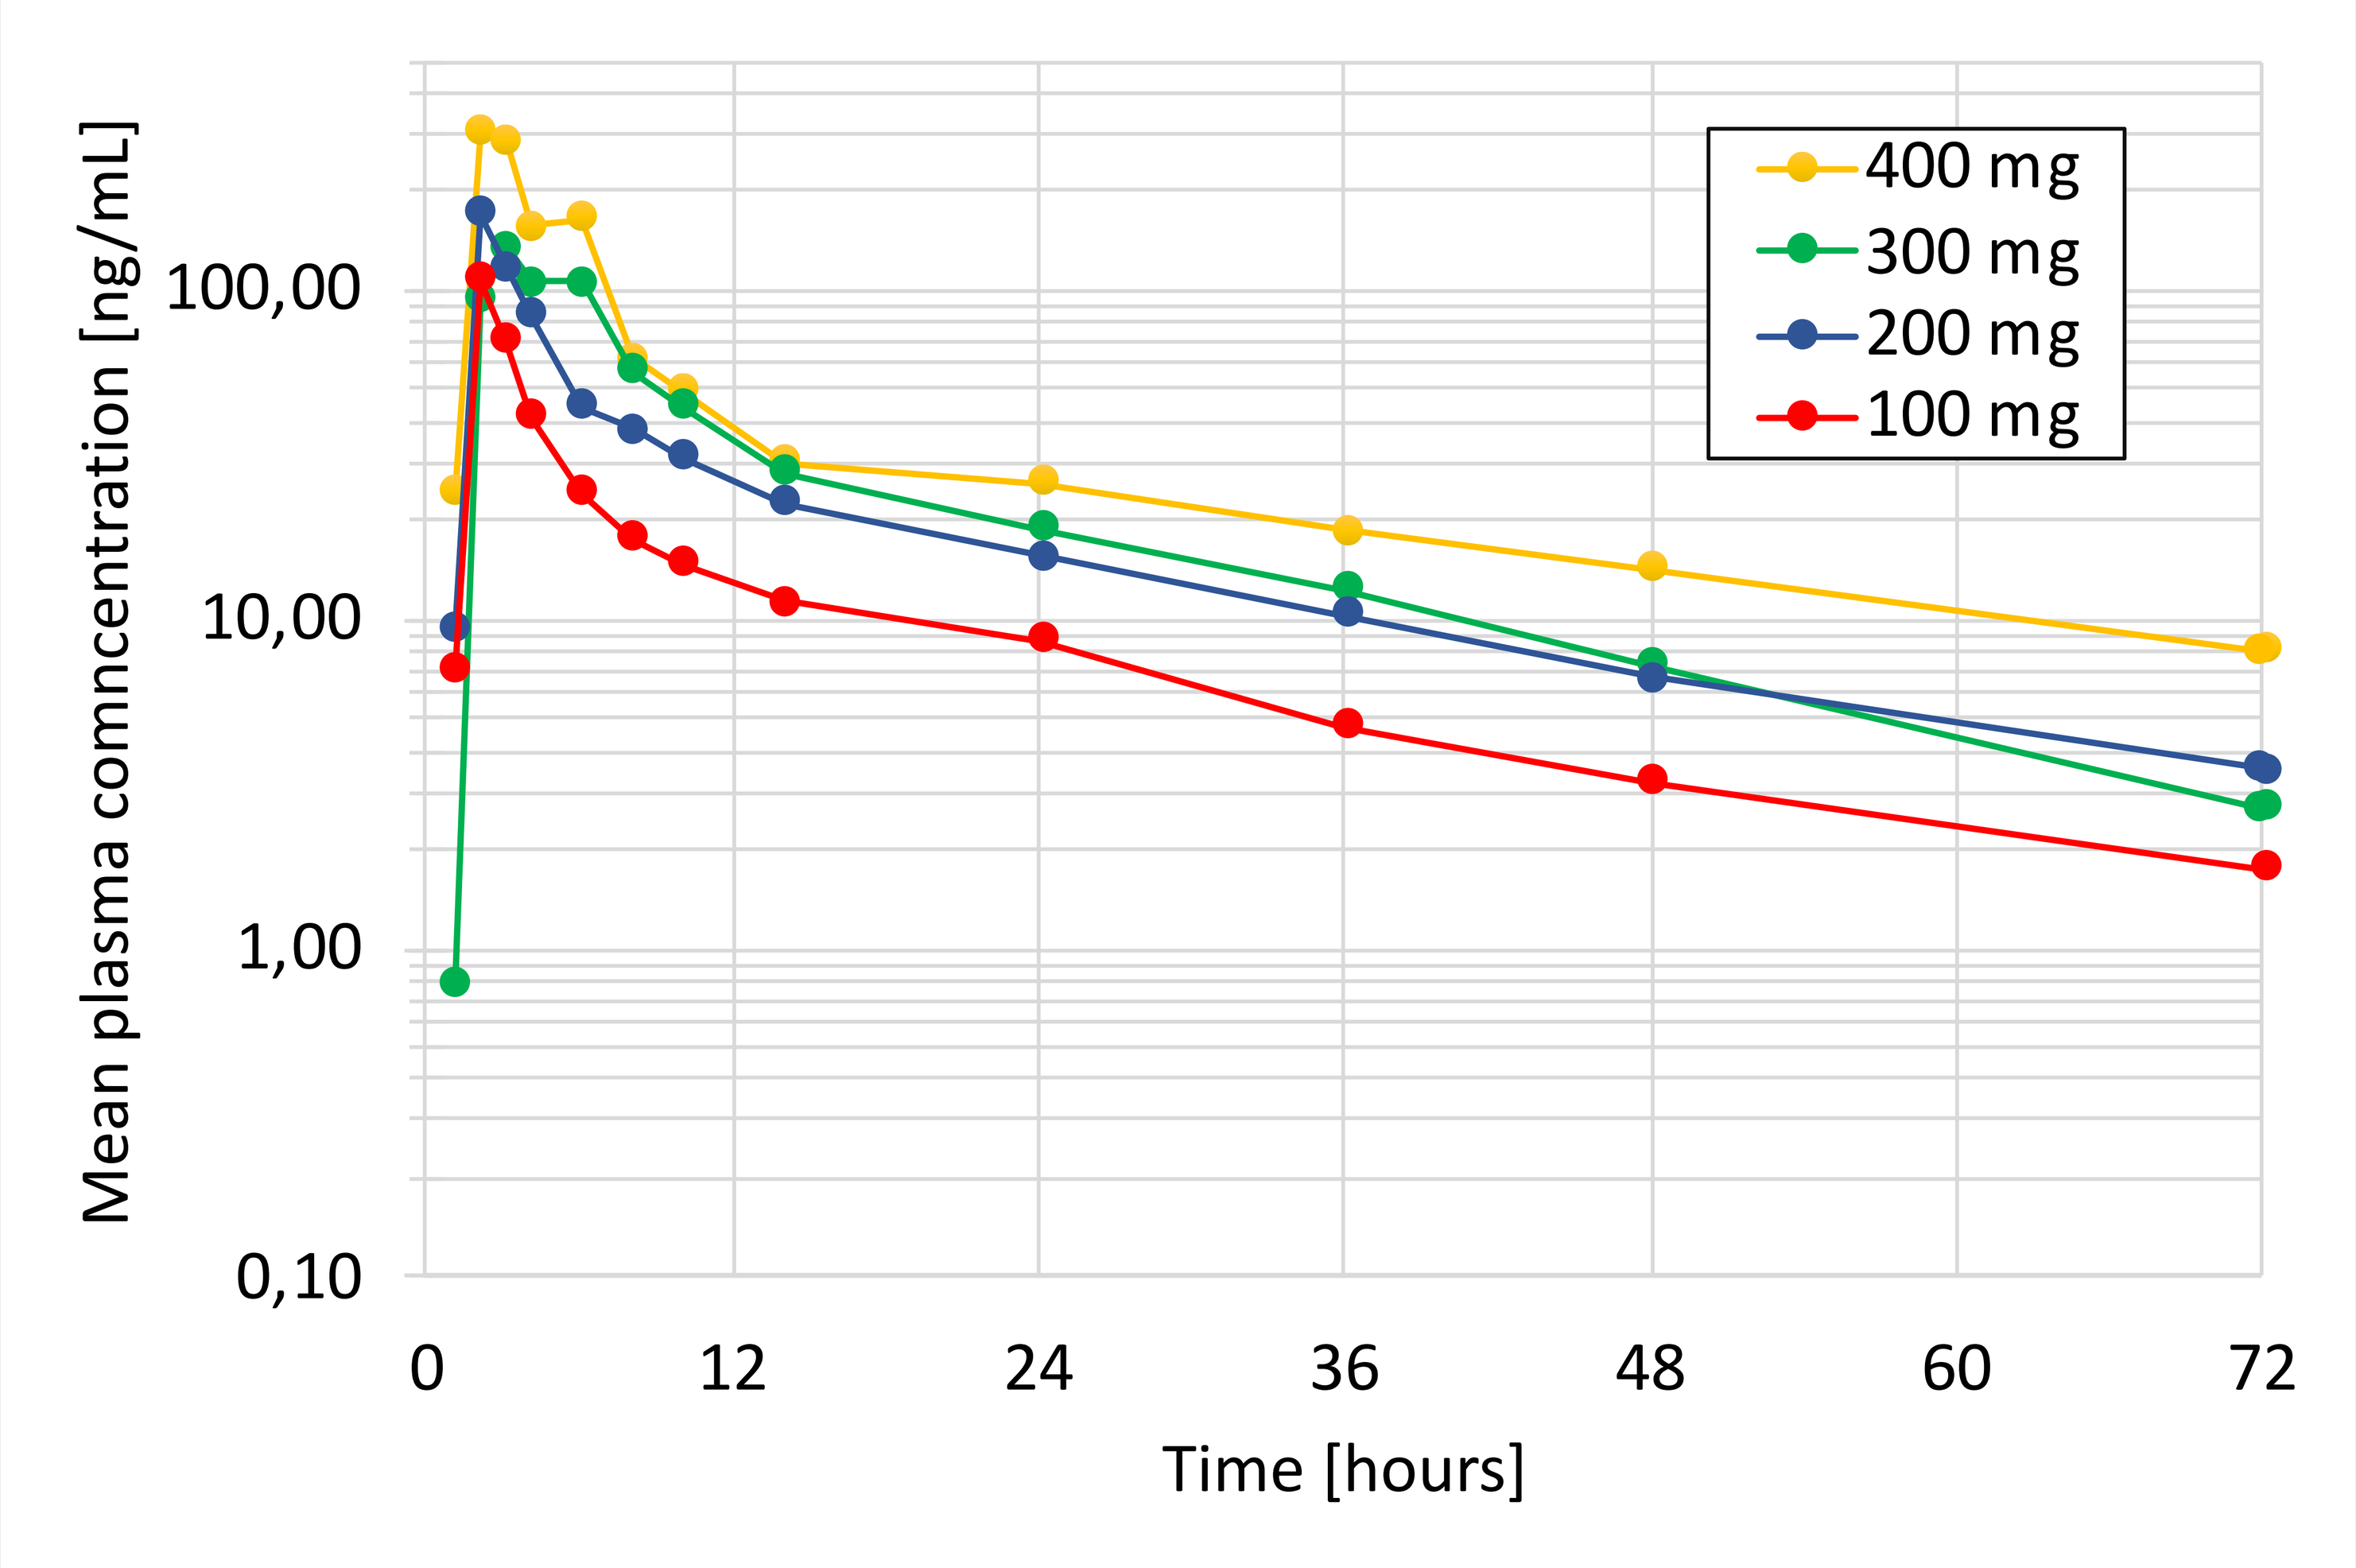
Mean plasma comncentration [ng/mL]
100,00
10,00
1,00
0,10
0
12
24
36
48
60
72
Time [hours]
400 mg
300 mg
200 mg
100 mg

Figure 2 IMU-935 plasma concentration over time

Results (cont.)

Safety evaluation of IMU-935 showed a very low incidence of related treatment-emergent adverse events (TEAEs). Monitoring of lab parameters, vital signs and 12-lead ECGs revealed no clinically relevant abnormalities.

Table 2 IMU-935 adverse event profile

MedDRA Preferred Term	Number (%) of subjects with TEAEs [Number of TEAEs reported]				
	100 mg IMU-935 or placebo (N=7)	200 mg IMU-935 or placebo (N=8)	300 mg IMU-935 or placebo (N=8)	400 mg IMU-935 or placebo (N=8)	Total Subjects (N=31)
Constipation		1 (13%) [1]		2 (25%) [2]	3 (10%) [3]
Headache		1 (13%) [1]	1 (13%) [1]		2 (6%) [2]
Abdominal distension		2 (25%) [2]			2 (6%) [2]
Diarrhea		1 (13%) [1]			1 (3%) [1]
Nausea		1 (13%) [1]			1 (3%) [1]
Abdominal pain				1 (13%) [1]	1 (3%) [1]
Dizziness				1 (13%) [1]	1 (3%) [1]
Limb injury		1 (13%) [1]			1 (3%) [1]
Lipase increased				1 (13%) [1]	1 (3%) [1]
Neutropenia	1 (14%) [1]				1 (3%) [1]
Somnolence	1 (14%) [1]				1 (3%) [1]

Results are presented in a blinded manner, where each treatment group includes subjects allocated to IMU-935 or placebo at a 3:1 ratio.

Conclusion

- ➢ IMU-935 has dose-linear pharmacokinetics and a plasma half-life that allows for once daily dosing.
- ➢ IMU-935 is safe and well tolerated with a benign adverse event profile.

Ongoing recruitment into this study will

1) characterize the multiple dose properties of IMU-935, and
2) provide data on the safety and activity of IMU-935 in psoriasis patients.

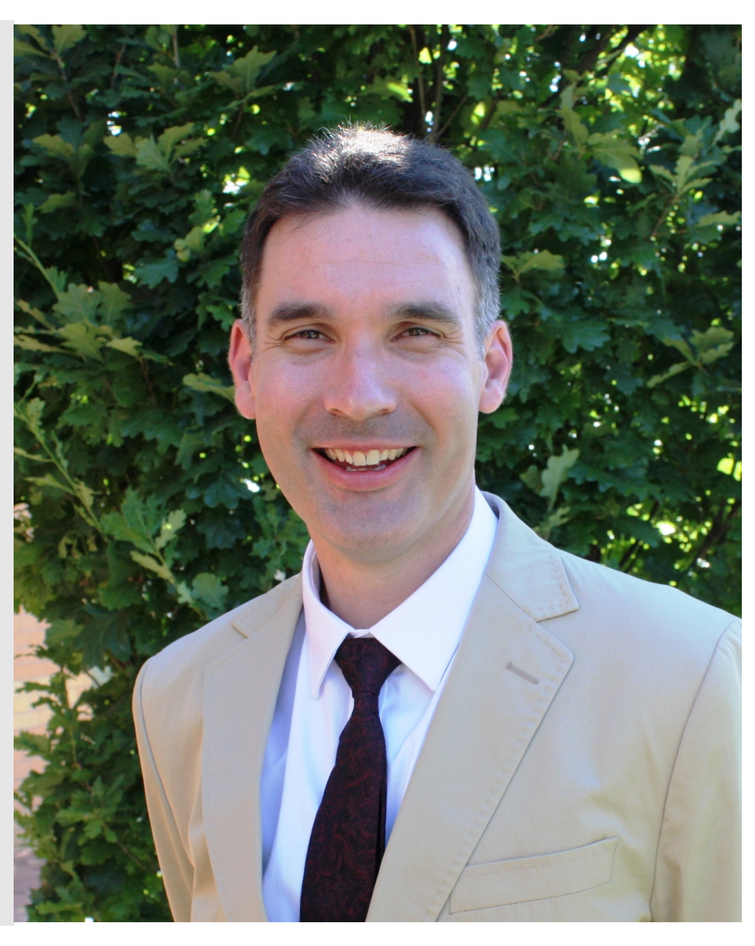

Royal Adelaide Hospital, SA Health
Thomas M Polasek
Clinical Pharmacology Registrar
Department of Clinical Pharmacology
Level 8E, 1 Port Road, Adelaide, South Australia, Australia, 5000

E-mail: tom.polasek@certara.com
Phone: +61 458 162715
Web: https://www.linkedin.com/in/thomas-polasek-378706b5/

Government of South Australia
SA Health

Immunic THERAPEUTICS