IMU-838, a developmental DHODH inhibitor in phase II for autoimmune disease, shows anti-SARS-CoV-2 and broad-spectrum antiviral efficacy *in vitro*

Friedrich Hahn[1], Christina Wangen[1], Sigrun Häge[1], Antonia Sophia Peter[1], Gerhard Dobler[2], Brett Hurst[3], Justin Julander[3], Jonas Fuchs[4], Zsolt Ruzsics[4], Klaus Überla[1], Hans-Martin Jäck[5], Roger G. Ptak[6], Andreas Muehler[7], Manfred Gröppel[7], Daniel Vitt[7], Evelyn Peelen[7], Hella Kohlhof[7,&], Manfred Marschall[1,&]

[1] Institute for Clinical and Molecular Virology, Friedrich-Alexander University of Erlangen-Nürnberg (FAU), Schlossgarten 4, 91054 Erlangen, Germany
[2] Institute of Microbiology, Bundeswehr, Neuherbergstraße 11, 80937 München, Germany
[3] Institute for Antiviral Research, Utah State University, 5600 Old Main Hill, Logan, UT 84322, USA
[4] Institute for Virology, University Medical Center/Universitätsklinikum Freiburg, Hermann-Herder Str 11, 79104 Freiburg, Germany
[5] Division of Immunology, Department of Medicine 3, FAU, Glückstraße 6, 91054 Erlangen, Germany
[6] Drug Development Division, Infectious Disease Research, Southern Research, 431 Aviation Way, Frederick, Maryland 21701, USA
[7] Immunic AG, Lochhamer Schlag 21, 82166 Gräfelfing, Germany
[&] Both authors contributed equally to this study

I IMU-838

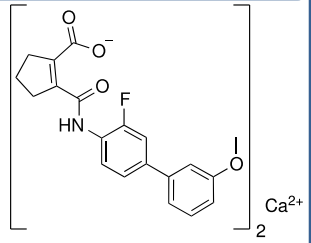
Ca2+
2

$C_{40}H_{34}CaF_2N_2O_8$
748.79 g/mol

IMU-838
- Inhibitor of the human dihydroorotate dehydrogenase (DHODH)
- Calcium salt of vidofludimus
- Investigational drug for the treatment of chronic inflammatory and autoimmune diseases

II IMU-838 reduces viral protein production and dsRNA synthesis

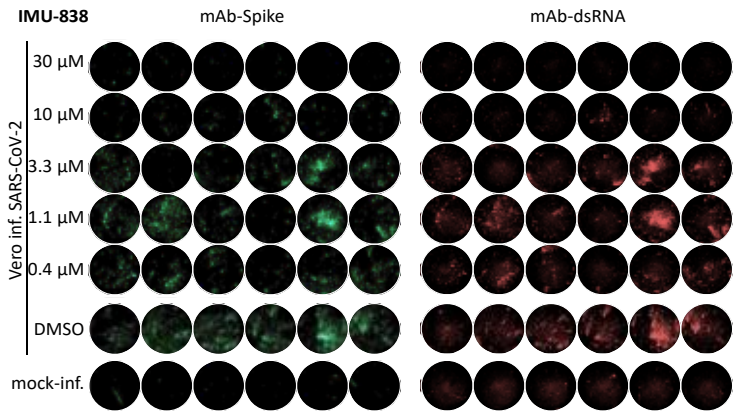
IMU-838
mAb-Spike
mAb-dsRNA
Vero inf. SARS-CoV-2
30 µM
10 µM
3.3 µM
1.1 µM
0.4 µM
DMSO
mock-inf.

Fig. 1: Parallel cultures of Vero cell were infected with SARS-CoV-2 at an MOI of 0.003 and treated with the indicated concentrations of IMU-838. At 30 h p.i., cells were fixed and stained with antibodies specific for the viral Spike protein and double-stranded-RNA (dsRNA).

- **IMU-838 inhibits viral Spike protein and dsRNA production in a concentration-dependent manner**

III IMU-838 inhibits SARS-CoV-2 virus production

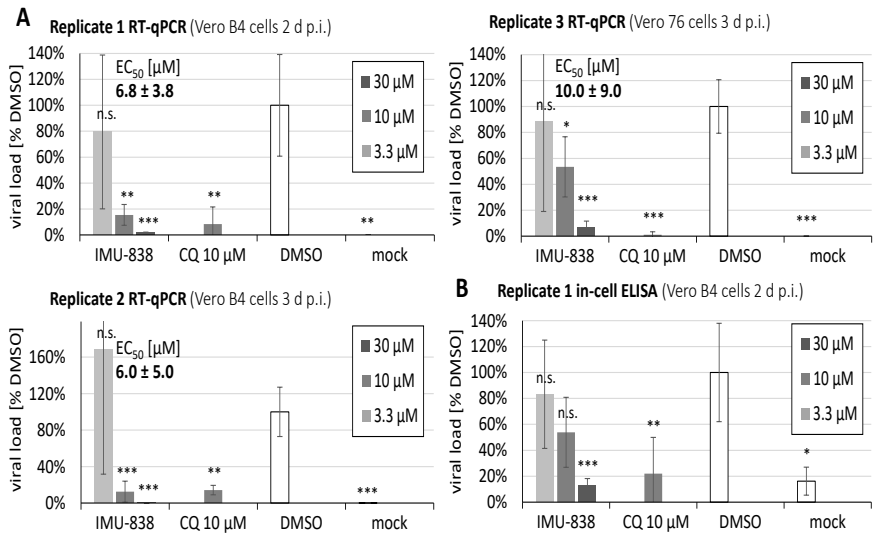
A
Replicate 1 RT-qPCR (Vero B4 cells 2 d p.i.)
EC50 [µM]
6.8 ± 3.8
Replicate 3 RT-qPCR (Vero 76 cells 3 d p.i.)
EC50 [µM]
10.0 ± 9.0
Replicate 2 RT-qPCR (Vero B4 cells 3 d p.i.)
EC50 [µM]
6.0 ± 5.0
B
Replicate 1 in-cell ELISA (Vero B4 cells 2 d p.i.)
viral load [% DMSO]
30 µM
10 µM
3.3 µM
IMU-838
CQ 10 µM
DMSO
mock

Fig. 2: Antiviral activity of IMU-838 determined by RT-qPCR quantitation of released viral genomes in supernatants from SARS-CoV-2-infected Vero cells treated with indicated concentrations of IMU-838 (A), and subsequently by in-cell ELISA using the remaining cell layers (B).

- **IMU-838 inhibits SARS-CoV-2 replication in Vero cells with a mean EC_{50} of 7.6 ± 5.8 µM**

IV Anti SARS-CoV-2 activity of IMU-838 in human cells

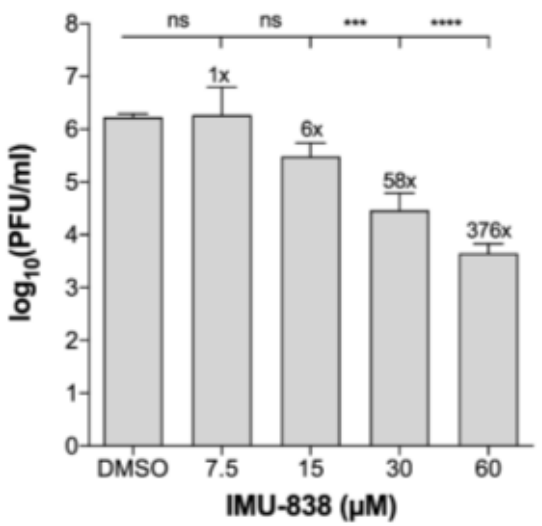
ns
ns

1x
6x
58x
376x
log10(PFU/ml)
DMSO
7.5
15
30
60
IMU-838 (µM)

Fig. 3: CaCo-2 cells were infected with SARS-CoV-2 at an MOI of 0.001 for 1.5 h and subsequently treated as with IMU-838 as indicated. After 72 h, viral titers were determined by plaque assay in Vero E6 cells.

- **IMU-838 exhibits anti-SARS-CoV-2 activity in human CaCo-2 cells**

V Combinatorial antiviral *in vitro* effect of IMU-838–RDV

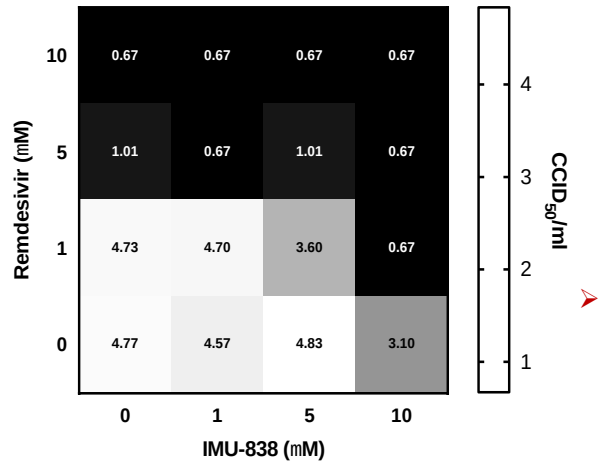
Remdesivir (mM)
IMU-838 (mM)
CCID50/ml
10: 0.67 0.67 0.67 0.67
5: 1.01 0.67 1.01 0.67
1: 4.73 4.70 3.60 0.67
0: 4.77 4.57 4.83 3.10
0 1 5 10

Fig. 4: Vero 76 cells infected with SARS-CoV-2 were cultured for 3 days with indicated concentrations of IMU-838 or RDV either alone or in combination. Depicted are the log-transformed viral yields determined by endpoint dilution assay 3 d p.i.

- **Effect of 5 µM of RDV combined with 1 µM of IMU-838 suggested highly promising potential of this drug combination *in vitro*.**

VI Anti-SARS-CoV-2 activity of IMU-CO3

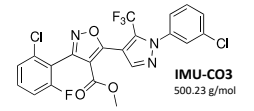
IMU-CO3
500.23 g/mol

IMU-CO3
- High-affinity back-up inhibitor of human DHODH

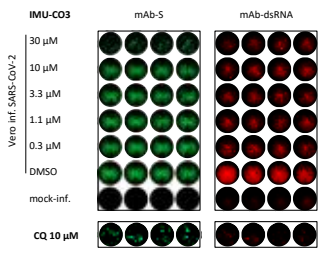
IMU-CO3
mAb-S
mAb-dsRNA
Vero inf. SARS-CoV-2
30 µM
10 µM
3.3 µM
1.1 µM
0.3 µM
DMSO
mock-inf.
CQ 10 µM

Anti-SARS-CoV-2 drug efficacy on Vero cells [µM]

	IMU-838	IMU-CO3
EC_{50}	7.6 ± 5.8	15.5 ± 4.6
CC_{50}	>100	>100
SI	>12.8	>6.5

	CQ	RDV
EC_{50}	2.7 ± 0.9	1.7 ± 1.0
CC_{50}	>100	>100
SI	>37	>59

Fig. 5: Antiviral activity of IMU-CO3 was determined by RT-qPCR using infectious supernatants of SARS-CoV-2-infected Vero cells and by in-cell ELISA using the remaining cell layers.

- **IMU-CO3 inhibits SARS-CoV-2 replication in Vero cells**

VII Anti-SARS-CoV-2 activity of IMU-838

Cells	SARS-CoV-2 isolate	MOI	Assay	Cmp	EC_{50} [µM]	CC_{50} [µM]	SI_{50}	EC_{90} [µM]	CC_{90} [µM]	SI_{90}
Vero 76 [b]	USA_WA1/2020	0.002	VYR	IMU-838	-	-	-	6.2 ±1.9	>100 [e]	>16
				TFNM	-	-	-	14	-	-
Vero 76 [c]	MUC-IMB-1	0.0002	RT-qPCR	IMU-838	10.0 ±9.0	88 ±4	>10.0	-	-	-
Vero B4 [c]	MUC-IMB-1	0.0002	RT-qPCR	IMU-838	6.8 ±3.8 (2 d) 6.0 ±5.0 (3 d)	>100	>14.7	-	-	-
CaCo-2 [d]	MUC-IMB-1	0.001	VYR	IMU-838	7.5-15	>60 [e]	>4-8	15-30	>60 [e]	>2-4

VIII IMU-838 *in vitro* activity against a panel of selected human pathogenic viruses

Virus [a]	Family	Cells [b]	Assay [c]	Drug [d]	EC_{50} [µM]	CC_{50} [µM]	SI
HCMV	Herpesviridae	HFF	GFP-based replication assay	Vido	7.4 ±0.1	>100	>13.5
				GCV	1.69 ±0.8	>15 [e]	>8.8
HCV	Flaviviridae	Huh7	Luc reporter assay	Vido	5.9 (4.5-7.7)	>30	>5.1
				rIFNα-2b [f]	0.21 (0.17-0.26)	2	>9.5
HIV-1	Retroviridae	Human PBMC	RT assay	Vido	2.1 (1.0-4.4)	>100	>47.5
				AZT	$0.38x10^{-3}$ ($0.20x10^{-3}$-$0.63x10^{-3}$)	>1	>2655
			p24 ELISA	Vido	1.3 (0.6-2.7)	>100	>76.1
				AZT	$0.27x10^{-3}$ ($0.10x10^{-3}$-$0.53x10^{-3}$)	>1	>3693

Results

- **DHODH inhibitors IMU-838 and back-up IMU-CO3 exerted anti-SARS-CoV-2 activity**
- **Antiviral activity is exerted by inhibition of viral protein production, double-strand RNA synthesis and release of infectious virus**
- **Antiviral efficacy in Vero cells was observed in a micromolar range**
- **Anti-SARS-CoV-2 activity was distinct from cytotoxic effects (CC_{50} >100 µM)**
- **The *in vitro* potency of IMU-838 was confirmed using several Vero lineages and human cells**
- **Combination of IMU-838 with remdesivir showed enhanced anti-SARS-CoV-2 activity**
- **Vidofludimus, the active determinant of IMU-838, exerted a broad-spectrum activity against a selection of major human pathogenic viruses**

Conclusion

- **These findings strongly suggest that developmental DHODH inhibitors represent promising candidates for use as anti-SARS-CoV-2 therapeutics.**

www.med.fau.de Funded by WSS MA/SBT 2018.121.1, Marohn-Stiftung Alz/Iko-Hahn/2019, DFG MA 1289/7-3 Hahn *et al.*, Viruses 2020, 12(12), 1394; PMID 33291455
Friedrich.Hahn@uk-erlangen.de